RACE trial

EBMT Clinical Trials Office

European Society
for Blood and Marrow Transplantation

RACE trial

A prospective Randomized multicenter study comparing horse Antithymocyte globuline (hATG) + Cyclosporine A (CsA) with or without Eltrombopag as front-line therapy for severe aplastic anaemia patients

A study by the Severe Aplastic Anaemia Working Party (SAAWP)

Principal Investigators: Antonio Risitano and Régis Peffault de Latour

Grant providers: Novartis and Pfizer

Trial objective

The primary objective of this trial is to investigate whether Eltrombopag added to standard immunosuppressive treatment increases the rate of early (at three months) complete response in untreated aplastic anaemia patients.

The RACE trial is a prospective open label randomised phase III trial aiming to change standard practice in severe aplastic anaemia (SAA).

	Treatment	Dose (units)	Treatment Period
Arm A (standard):	ATGAM	40 mg/kg/day, iv	Day 1, 2, 3 and 4
	Cyclosporine A	5 mg/kg/day, po	Day 1-365
Arm B: arm A + Investigational drug	Eltrombopag	150 mg every 24 h, po	Day 14-90 (or 14-180)

Primary endpoint: **rate of Complete Response (CR) at 3 months** since start of treatment in naive SAA patients.

CR is defined as: haemoglobin>10 g/dL, absolute neutrophil count>1,000/µL and platelets>100,000 µL

Baseline characteristics RACE subjects

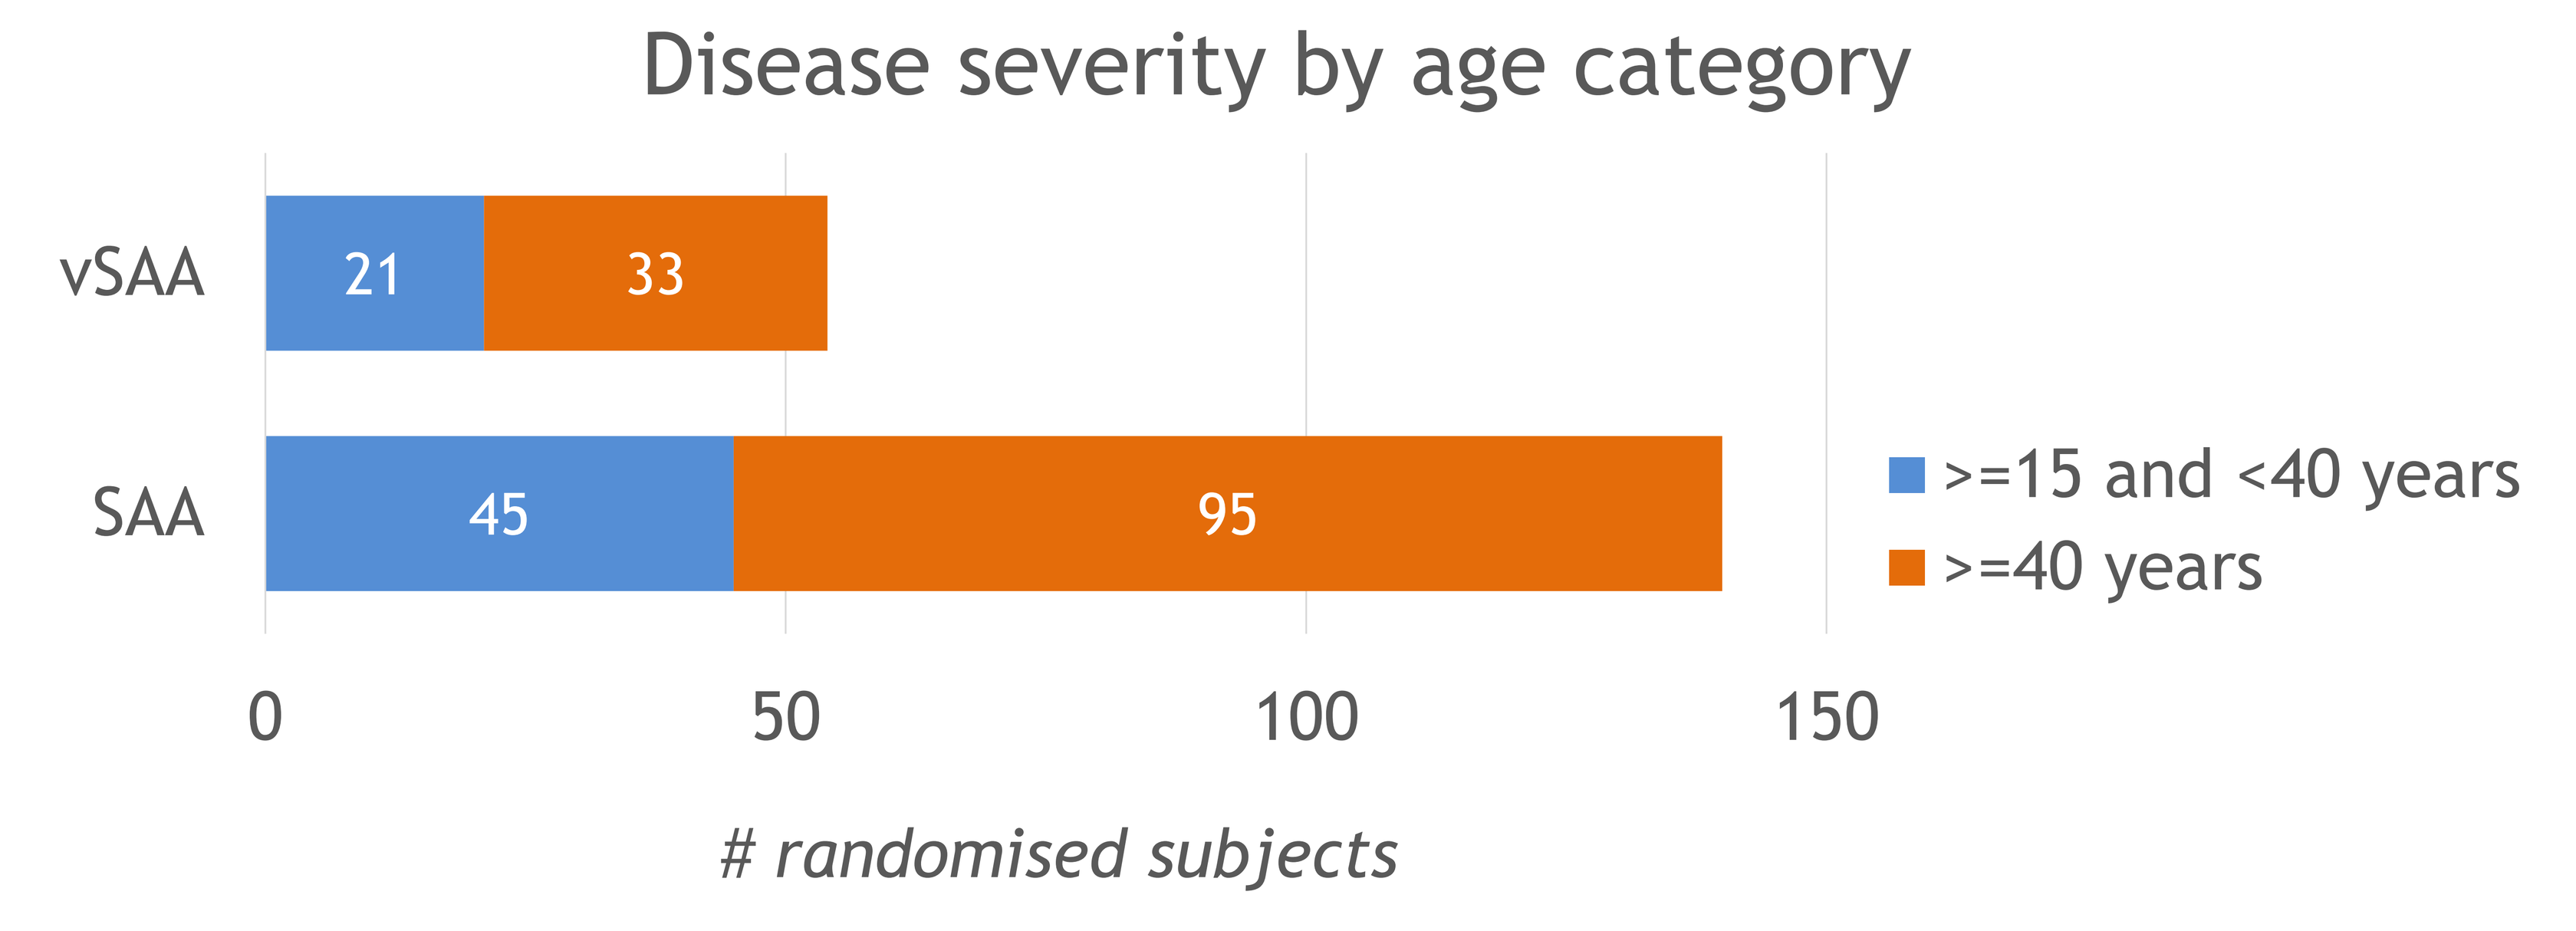
Disease severity by age category
vSAA
SAA
21
33
45
95
0
50
100
150
>=15 and <40 years
>=40 years
randomised subjects

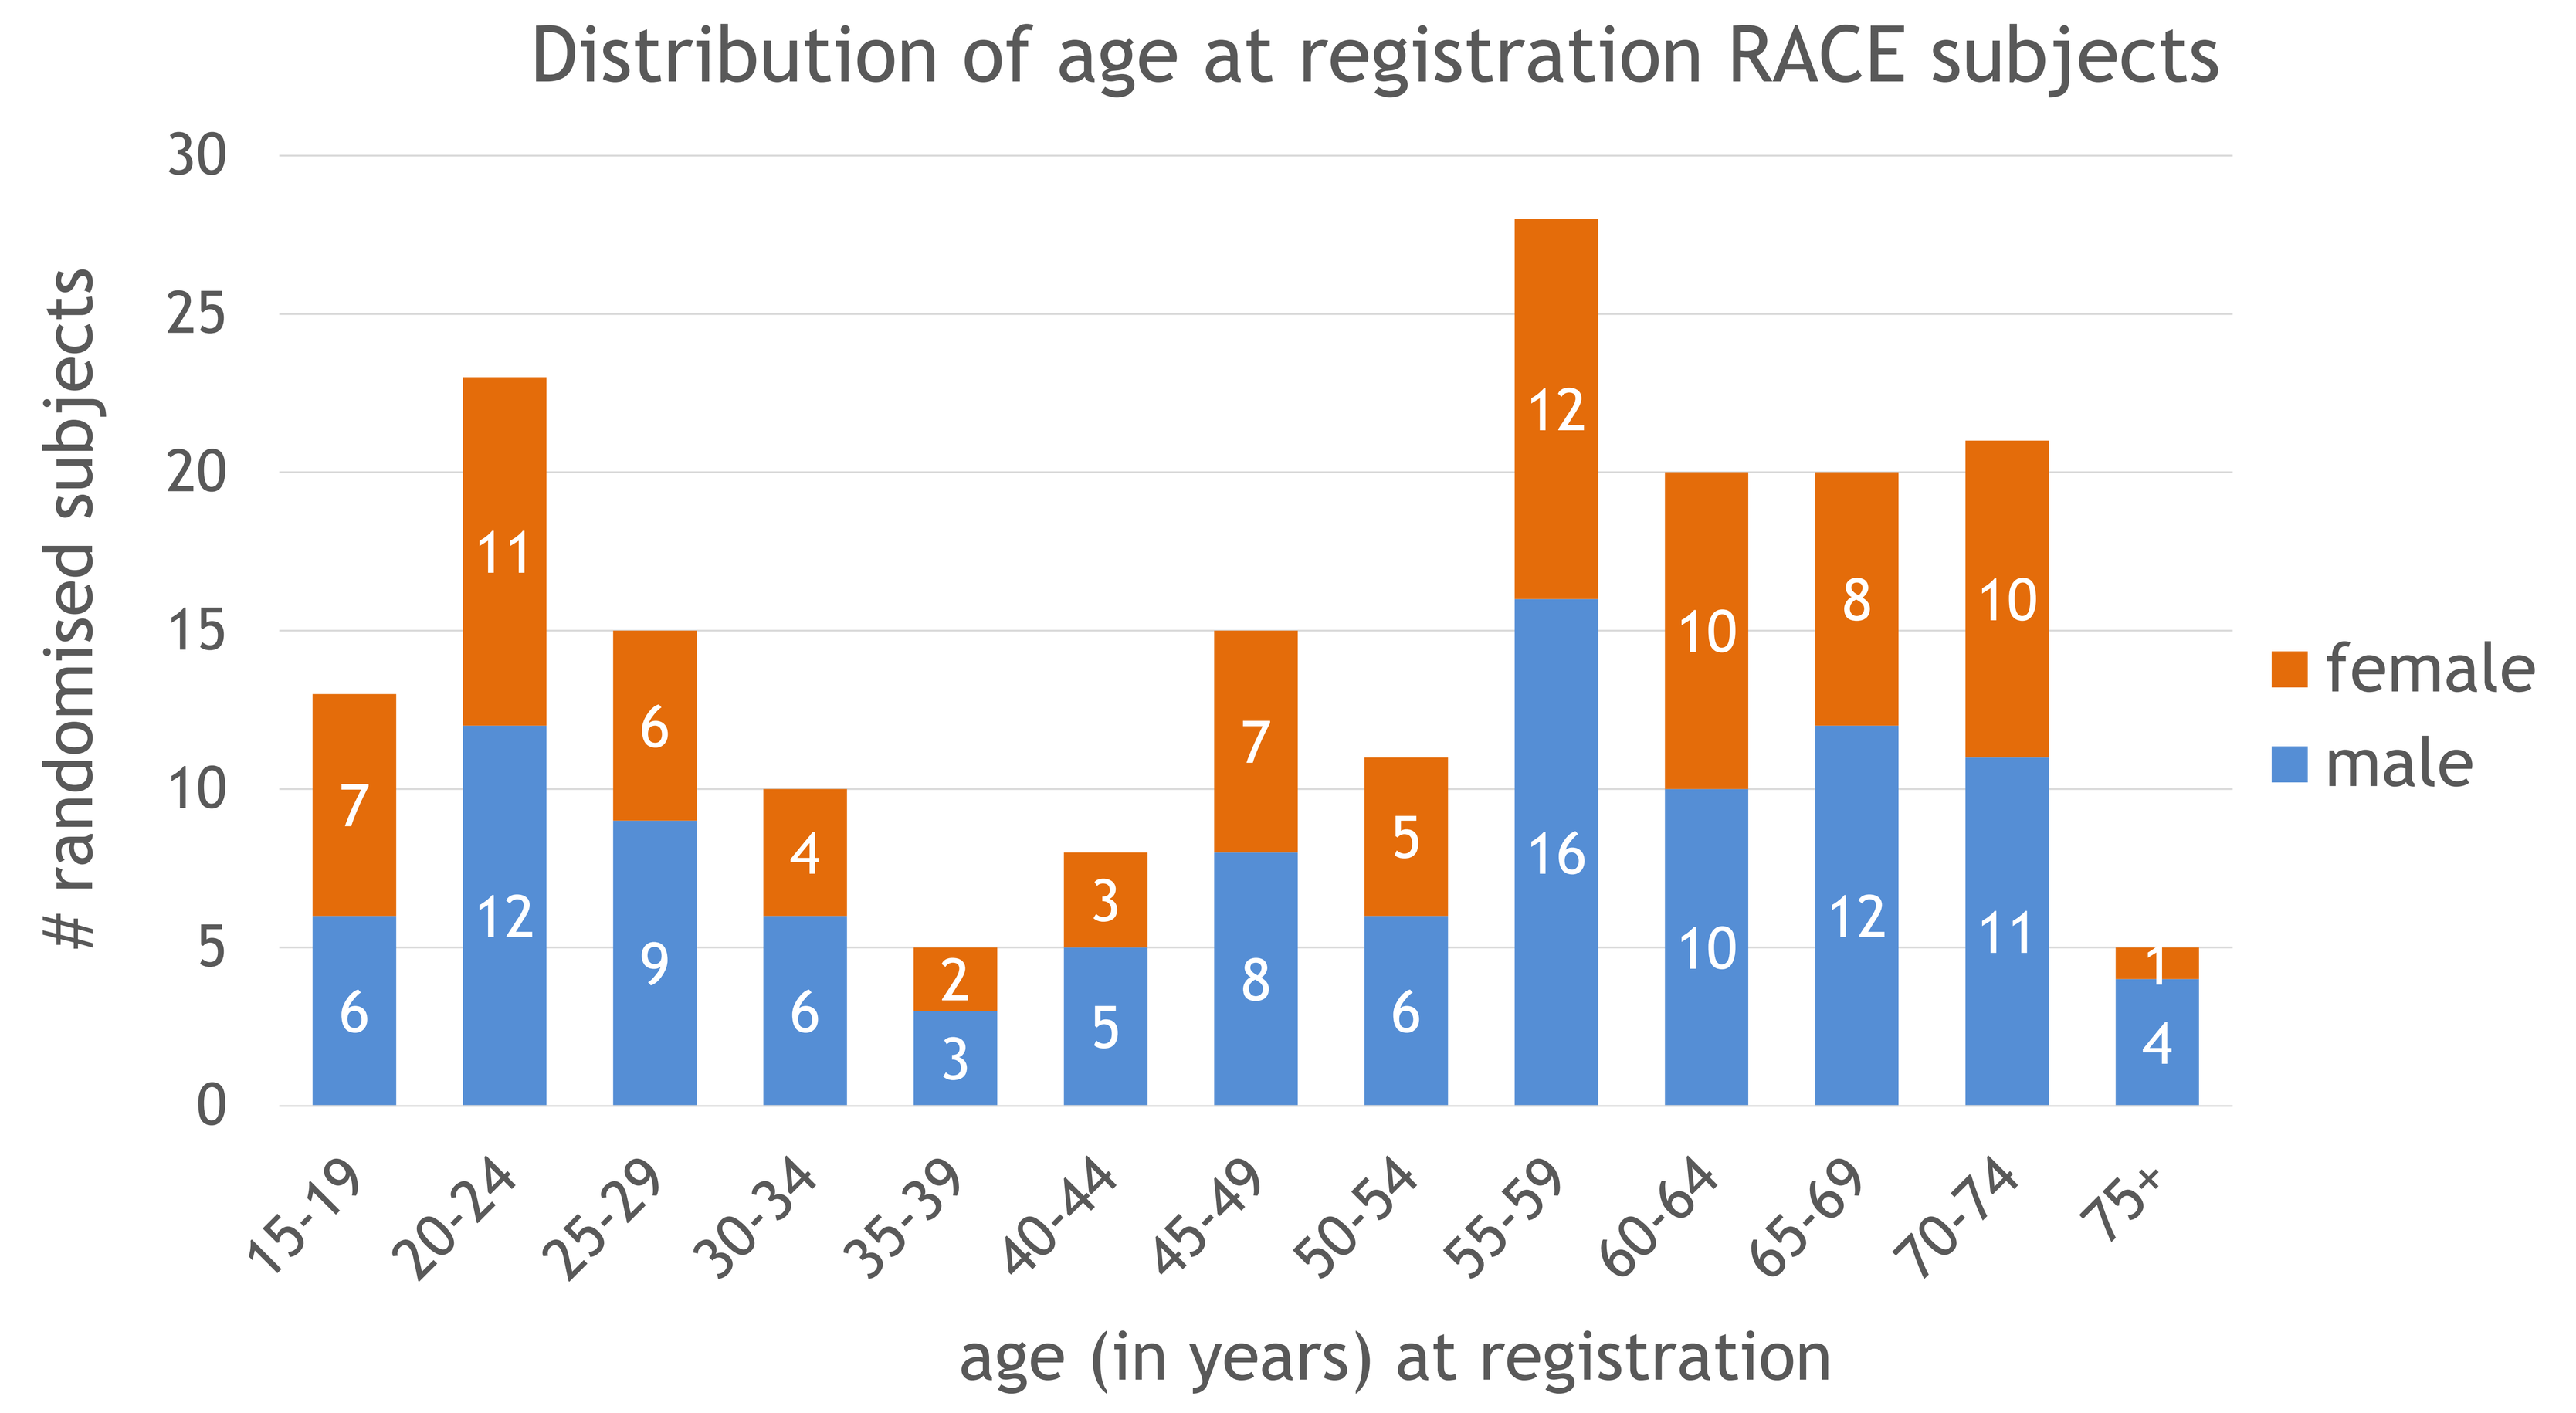
Distribution of age at registration RACE subjects
randomised subjects
30
25
20
15
10
5
0
15-19
20-24
25-29
30-34
35-39
40-44
45-49
50-54
55-59
60-64
65-69
70-74
75+
female
male
age (in years) at registration

Thank you!

Congratulations to all involved: the recruitment target of 200 subjects is almost reached! We look forward to recruiting the last patients and shortly thereafter publishing the first results of this landmark trial. We thank you for your excellent work and support!

Recruitment status (data on 4 March 2019)

Country	N° of open sites	N° of enrolling sites	N° of randomised subjects
France	7	7	80
Italy	6	4	27
Netherlands	4	4	26
Spain	4	4	11
Switzerland	3	1	7
United Kingdom	4	4	43
Total	**28**	**24**	**194**

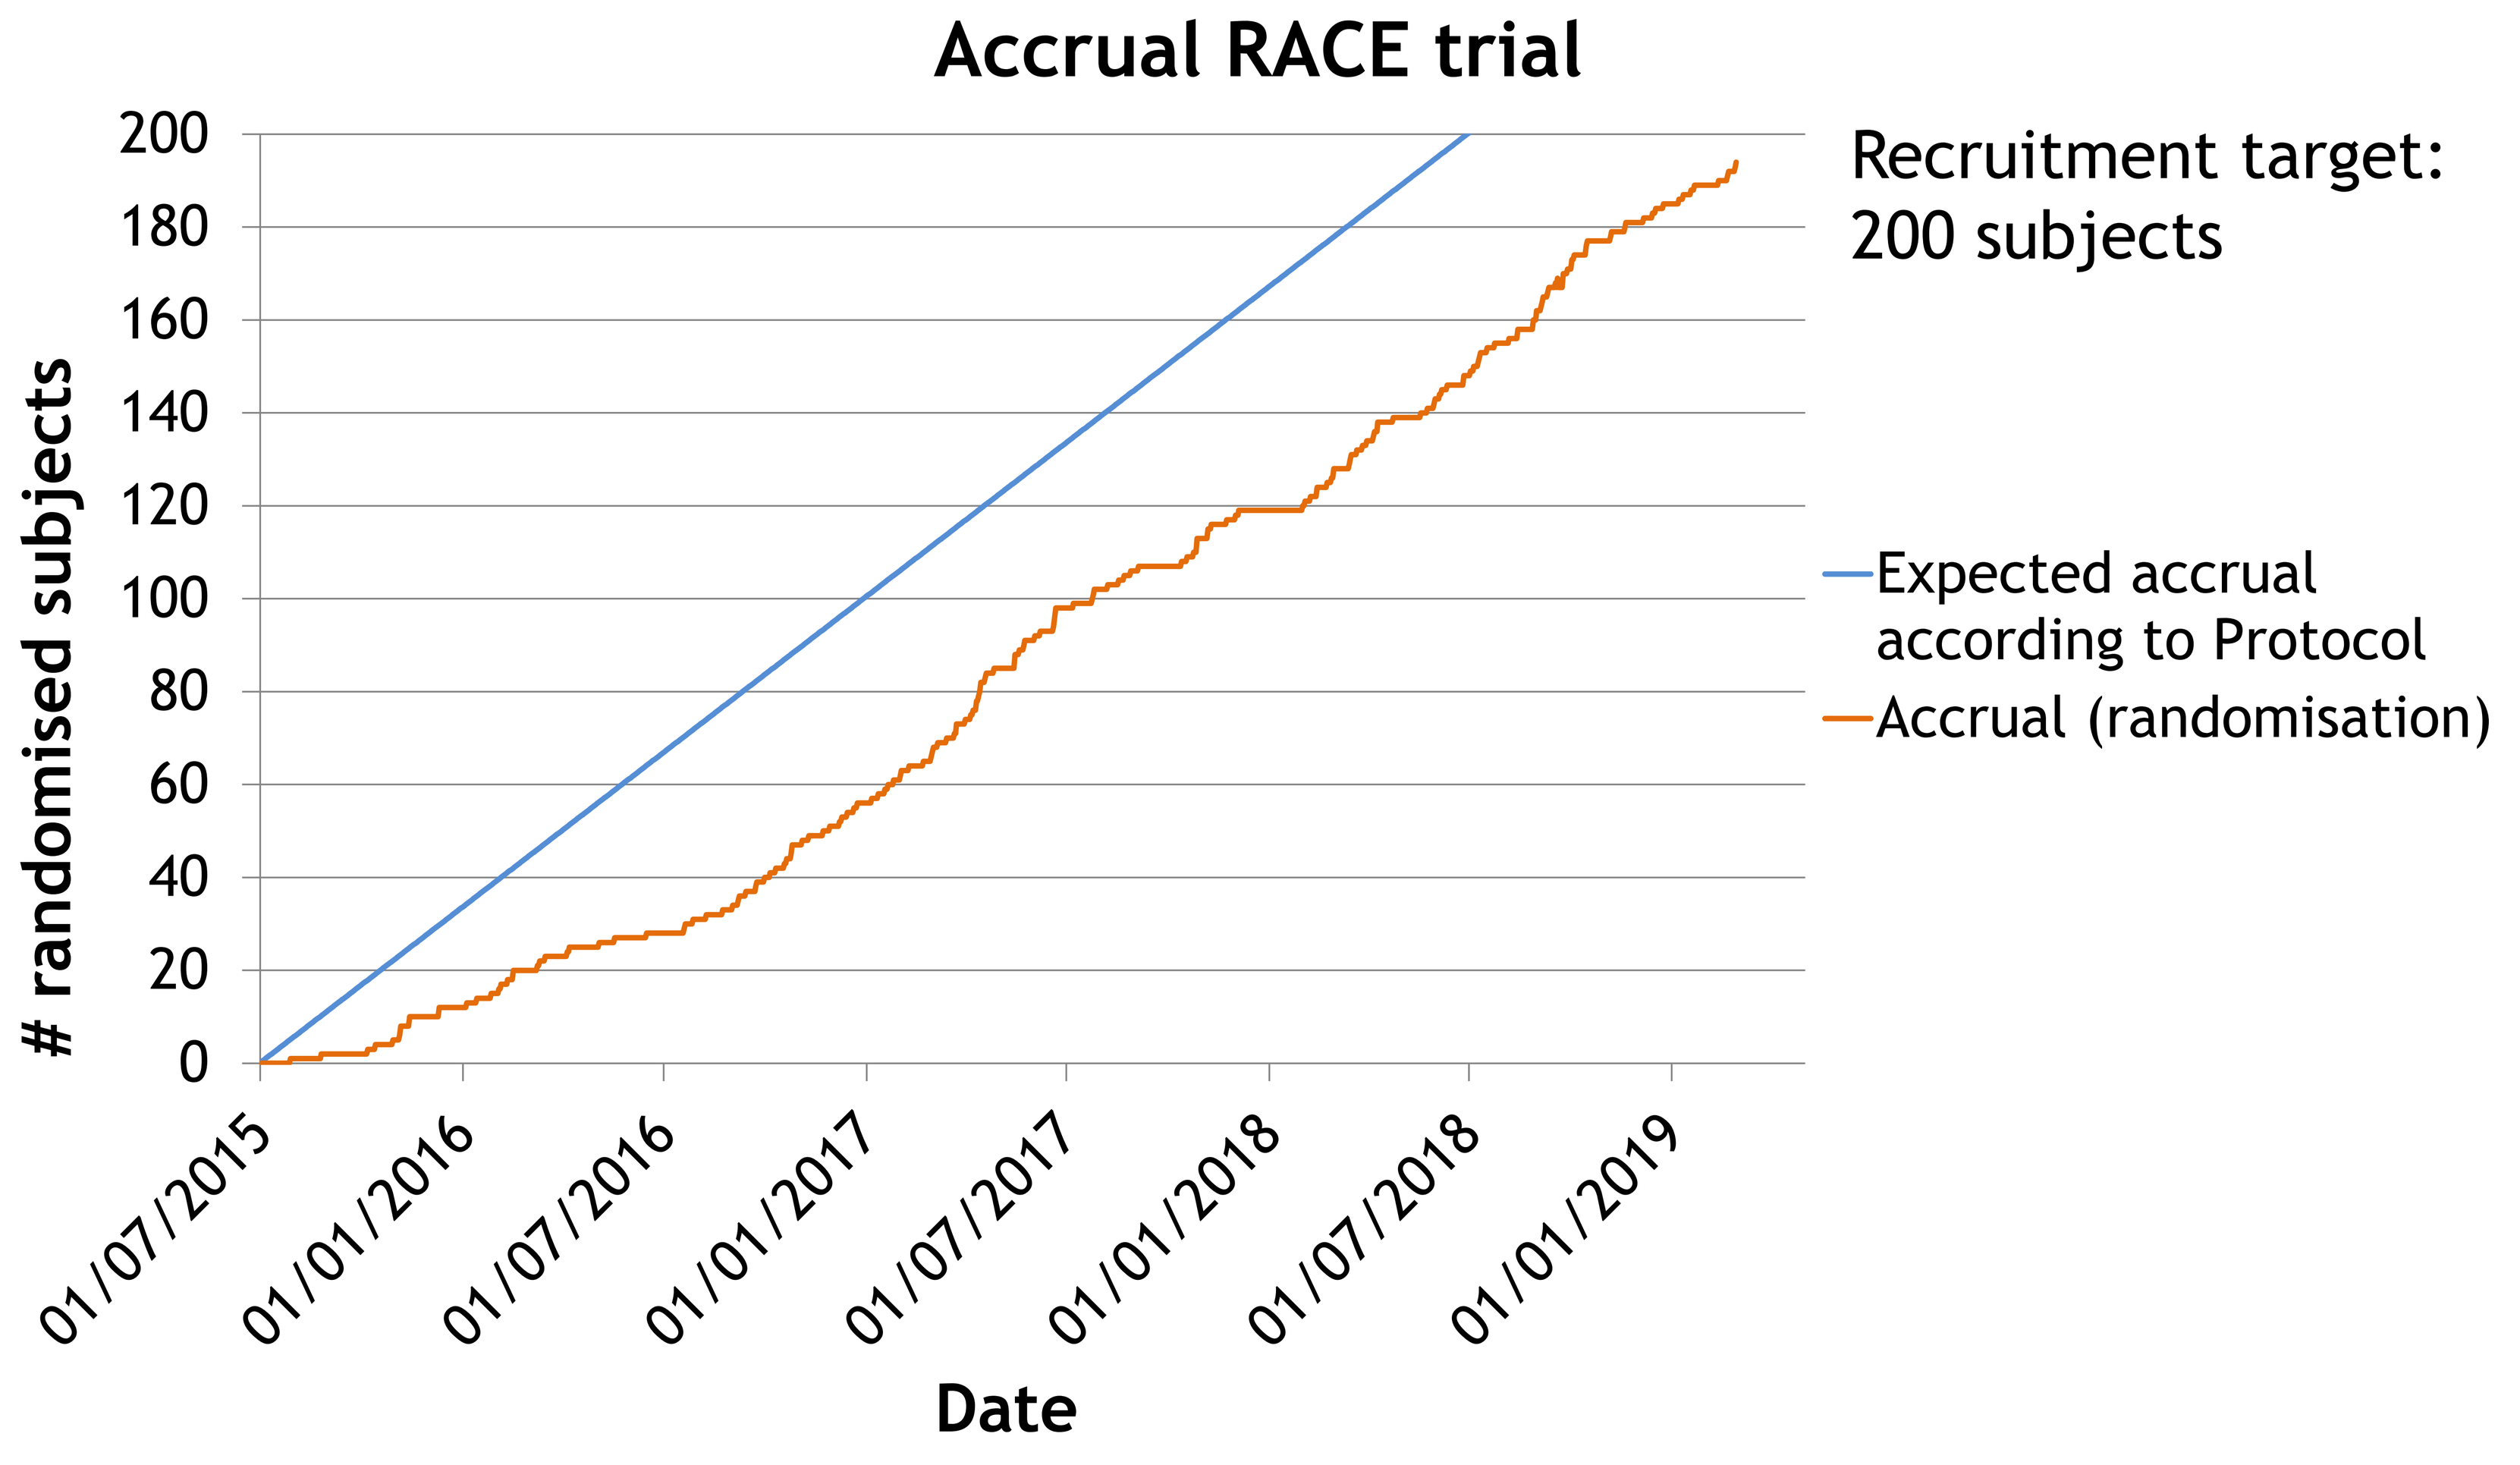
Accrual RACE trial
randomised subjects
200
180
160
140
120
100
80
60
40
20
0
01/07/2015
01/01/2016
01/07/2016
01/01/2017
01/07/2017
01/01/2018
01/07/2018
01/01/2019
Date
Recruitment target:
200 subjects
Expected accrual according to Protocol
Accrual (randomisation)

RACE-ing forward

• Expected last patient in:	April 2019
• Last patient 3m FU visit:	July 2019
• Last patient last visit:	April 2021
• First abstract (primary endpoint 3m FU):	ASH 2019
• Abstract presentation:	ASH 2021

Contact

Questions about RACE?

Contact Sofie Terwel at: +31 71 526 1183 or RACEtrial@lumc.nl

New Clinical Trial or Non-Interventional Study Proposals?

Contact Marianne Mol (CTO manager) at: +31 71 526 3953 or marianne.mol@ebmt.org

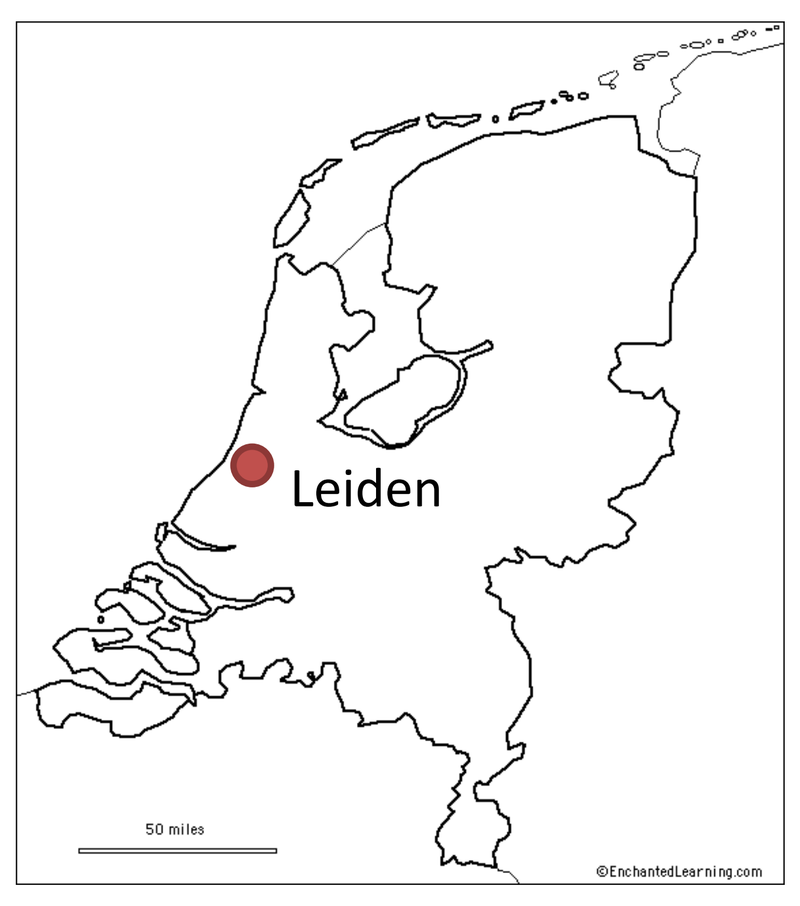
Leiden

EBMT Clinical Trials Office Leiden

Rijnsburgerweg 10
2333 AA Leiden
The Netherlands

Tel: +31 (0)71 526 1183
Fax: +31 (0)71 526 6185

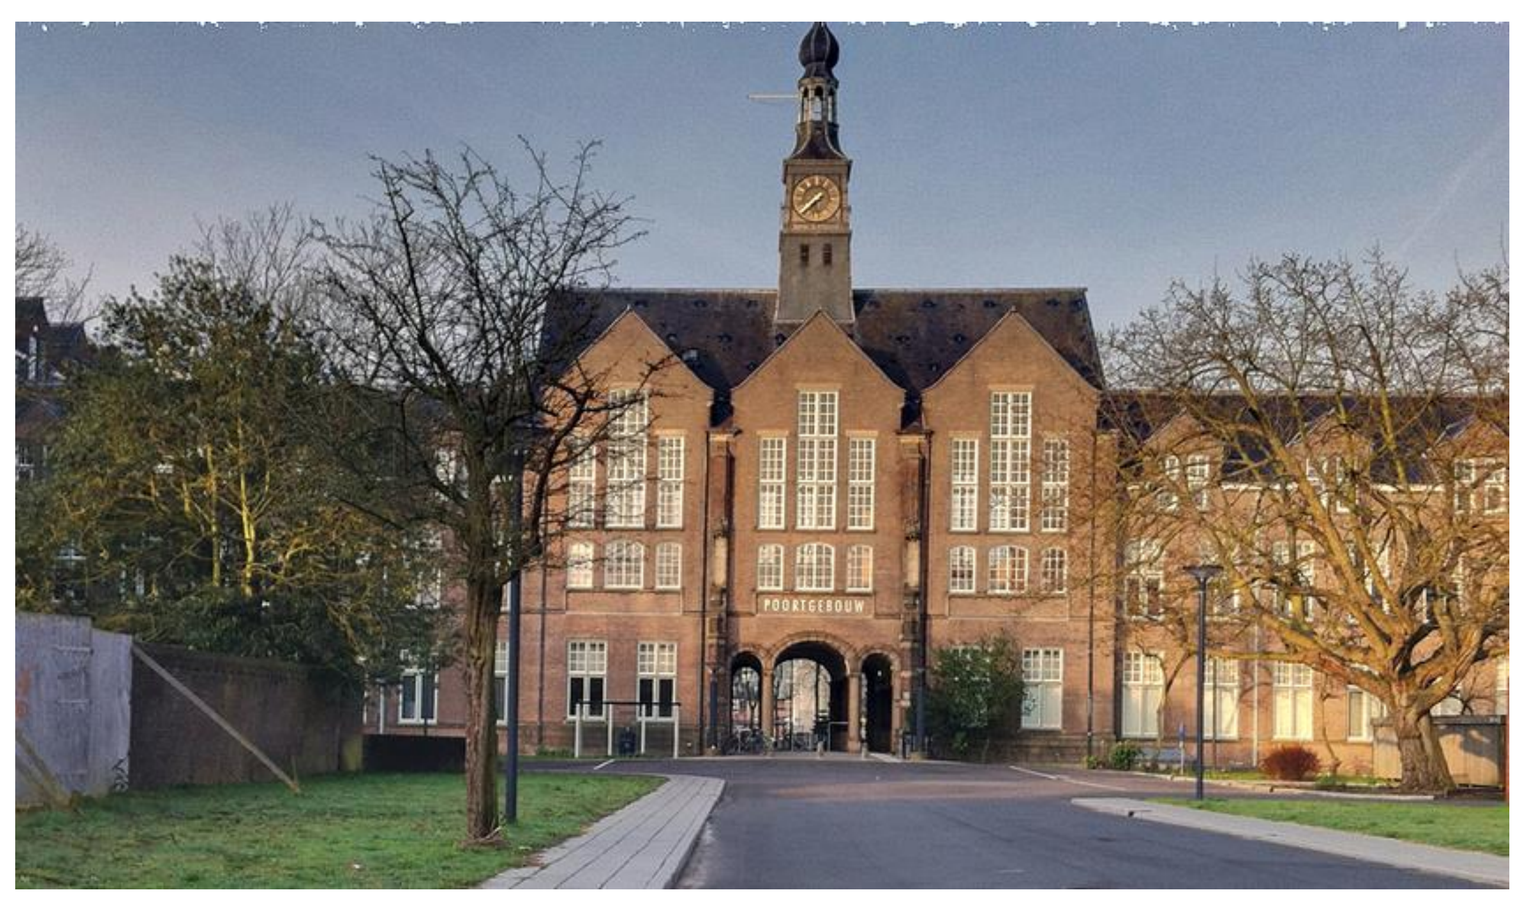

1

www.ebmt.org #EBMT19