EBMT
European Society
for Blood and Marrow Transplantation

Chronic Malignancies Working Party

MPN & Myelodysplastic Syndrome

Activities of WP and Subcommittees

Chair: Nicolaus Kröger, Secretary: Stefan Schönland

Subcommittee Myelodysplastic Syndrome

Chair: Marie Robin, Vice–Chair: Theo de Witte

Numbers of alloHCTs for MDS/sAL per year reported to the EBMT are increasing.

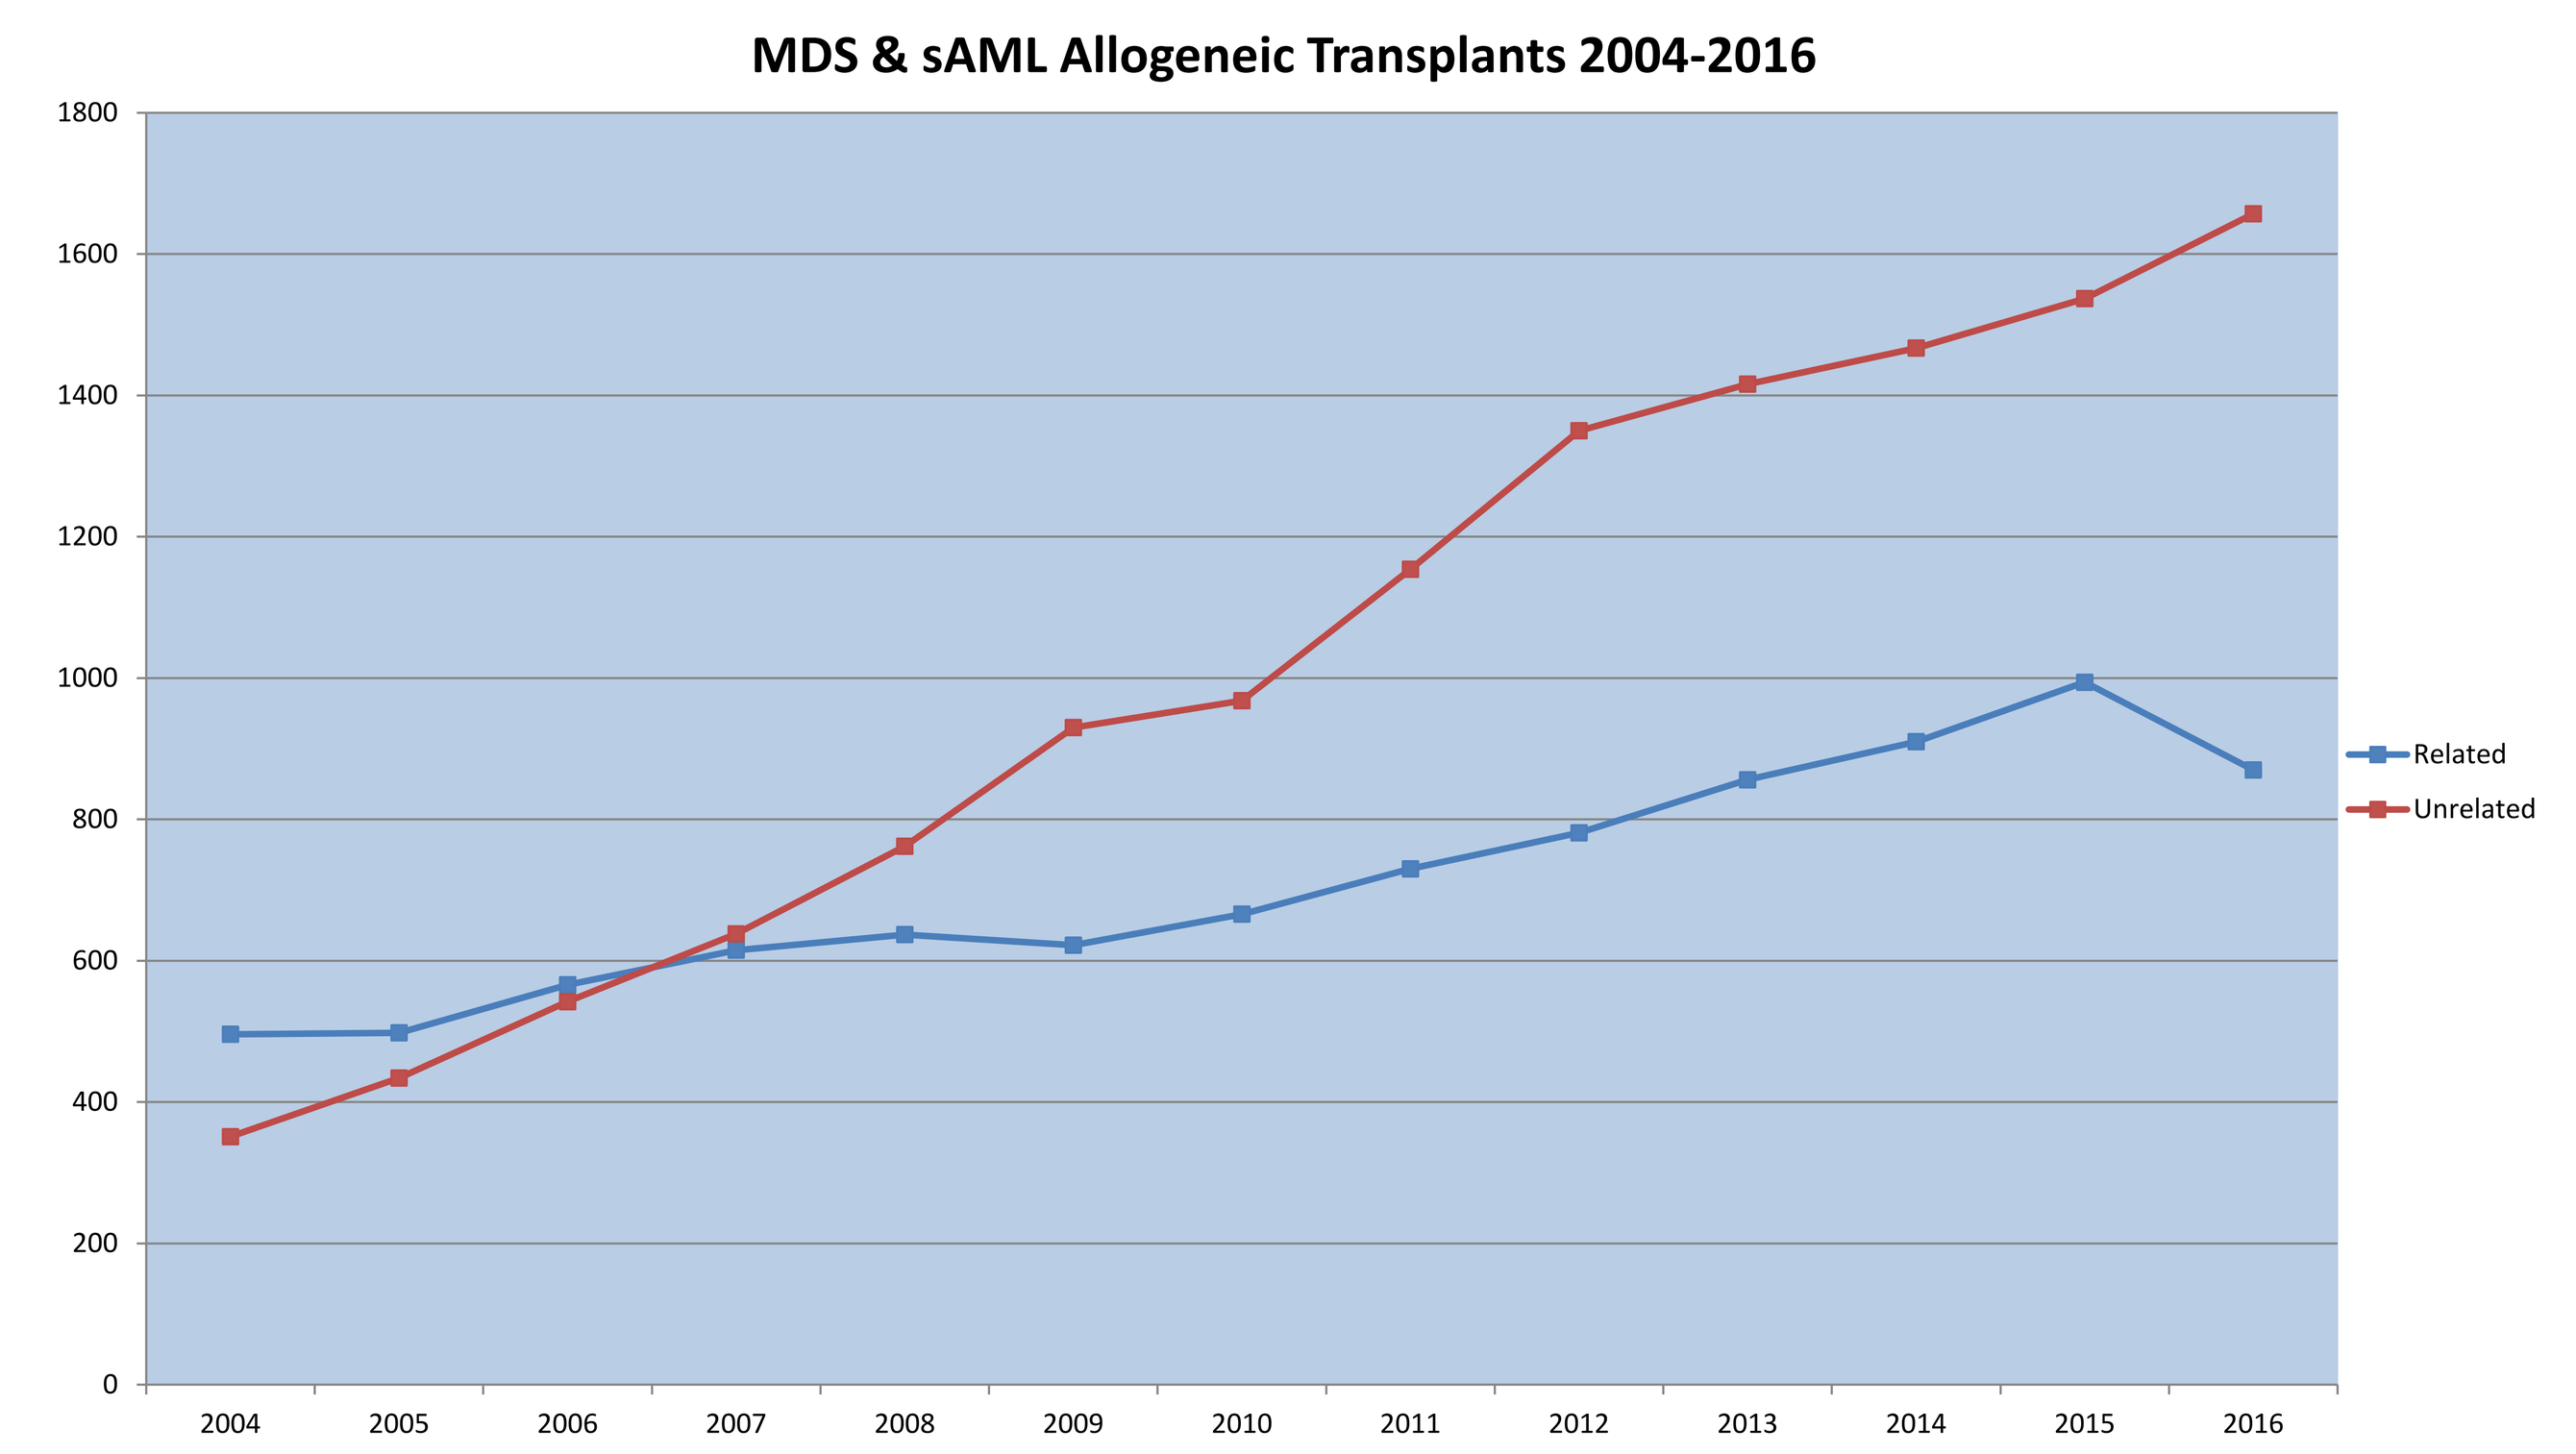
MDS & sAML Allogeneic Transplants 2004-2016
Related
Unrelated
0
200
400
600
800
1000
1200
1400
1600
1800
2004 2005 2006 2007 2008 2009 2010 2011 2012 2013 2014 2015 2016

Recommendations for the use of HSCT in MDS

Blood. 2017 Mar 30;129(13):1753-1762

An initiative to refine and update the general international guidelines for patients with MDS who are potential candidates for HSCT.

Using the information from the general MDS guidelines, an expert task force developed HSCT scenarios for patients with MDS, which were evaluated and discussed by a panel of experts during several consensus meetings. T. de Witte et al.

Recently accepted in JCO (35:2157)

Dose-reduced vs. standard conditioning followed by allogeneic stem cell transplantation for patients with myelodysplastic syndrome: A prospective randomized phase III study of the EBMT (RICMAC-Trial). N. Kröger et al.

EBMT 2018 presentations from the MDS subcommittee

Molecular genetics and transplant outcome in MDS
Marie Robin
Monday March 19th, CMWP session, 09:10-09:30, Room 3A

Long term outcome of patients with MDS surviving allogeneic stem cell transplantation
Johannes Schetelig
Monday March 19th, CMWP session, 10:10-10:30, Room 3A

Comparison of sequential versus myeloablative, reduced intensity and non-myeoloablative conditioning for patients with myelodysplastic syndrome
Victoria Potter et al.
Tuesday March 20th, 17:20-17:30, Auditorium VI

Comparative outcomes for matched and mismatched (haplo) family donors for myelodysplastic syndromes
Kavita Raj et al.
Wednesday March 21st, 11:20-11:30, Auditorium III

Posters
Tuesday March 20th, Poster Area

Poster B243 **External validation of scores predicting non-relapse mortality in patients with myelodysplastic syndrome.**
Martin Carre et al.

Poster B244 **Impact of in-vivo T-cell depletion in patients with myelodysplastic syndromes undergoing allogeneic HSCT**
Edouard Forcade et al

Other papers recently published (2017)

Haploidentical transplant in patients with myelodysplastic syndrome.
M. Robin et al *Blood Adv. 2017 Sep 27;1(22):1876-1883.*

Outcome after relapse of myelodysplastic syndrome and secondary acute myeloid leukemia after allogeneic stem cell transplantation: a retrospective registry analysis on 698 patients by the Chronic Malignancies Working Party of European Society of Blood and Marrow Transplantation.
C. Schmid et al *Haematologica 2017 Nov 3.*

Allogeneic Stem Cell Transplantation for Myelodysplastic Syndrome Patients with a 5q Deletion.
L. Garderet et al *Biol Blood Marrow Transplant. 2017 Nov 28.*

Validation of the revised IPSS at transplant in patients with myelodysplastic syndrome/transformed acute myelogenous leukemia receiving allogeneic stem cell transplantation: a retrospective analysis of the EBMT chronic malignancies working party.
C. Scheid et al *Bone Marrow Transplant. 2017 Sep 11.*

Long-term follow-up of a retrospective comparison of reduced-intensity conditioning and conventional high-dose conditioning for allogeneic transplantation from matched related donors in myelodysplastic syndromes.
R. Martino et al *Bone Marrow Transplant. 2017 Aug;52(8):1107-1112.*

Allogeneic Stem Cell Transplantation for Patients Age ≥ 70 Years with Myelodysplastic Syndrome: A Retrospective Study of the MDS Subcommittee of the Chronic Malignancies Working Party of the EBMT.
S. Heidenreich et al *Biol Blood Marrow Transplant. 2017 Jan;23(1):44-52.*

Allogeneic haematopoietic stem cell transplant in patients with lower risk myelodysplastic syndrome: a retrospective analysis on behalf of the Chronic Malignancy Working Party of the EBMT.
M. Robin et al *Bone Marrow Transplant. 2017 Jul;52(7):1081.*

Subcommittee Myeloproliferative Neoplasms

Chair: Yves Chalandon, Vice–Chair: Eduardo Olavarria

Numbers of alloHSCT for CML have decreased over time since introduction of TKI, but there is still a need for alloHSCT in this disease. In the contrary, number of alloHSCT for MPN per year reported to the EBMT are increasing.

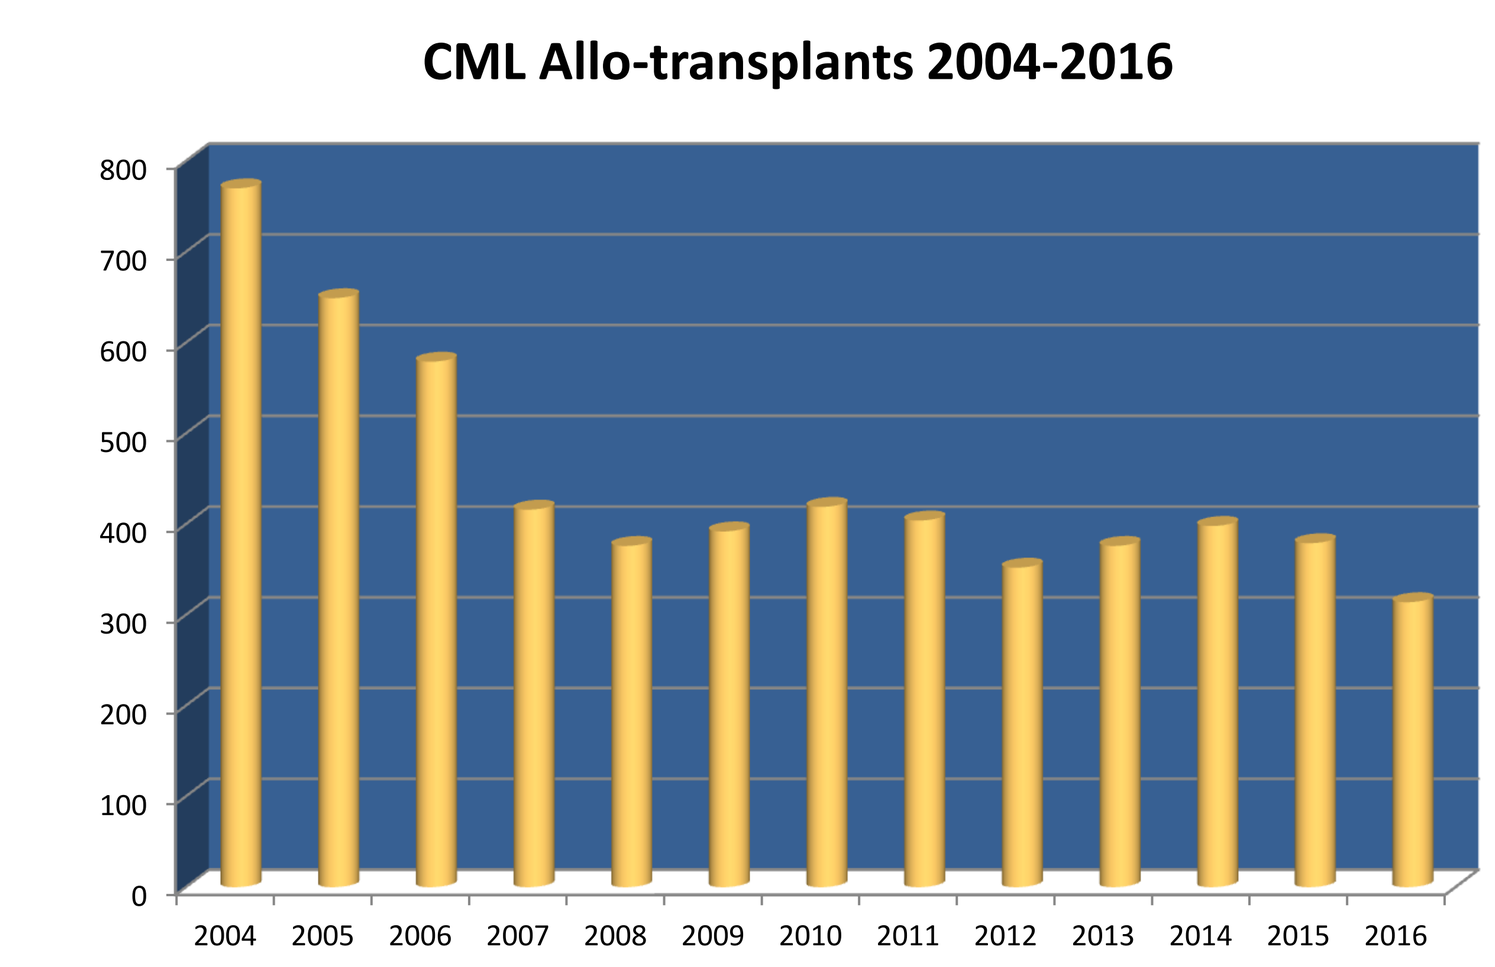
CML Allo-transplants 2004-2016
0
100
200
300
400
500
600
700
800
2004 2005 2006 2007 2008 2009 2010 2011 2012 2013 2014 2015 2016

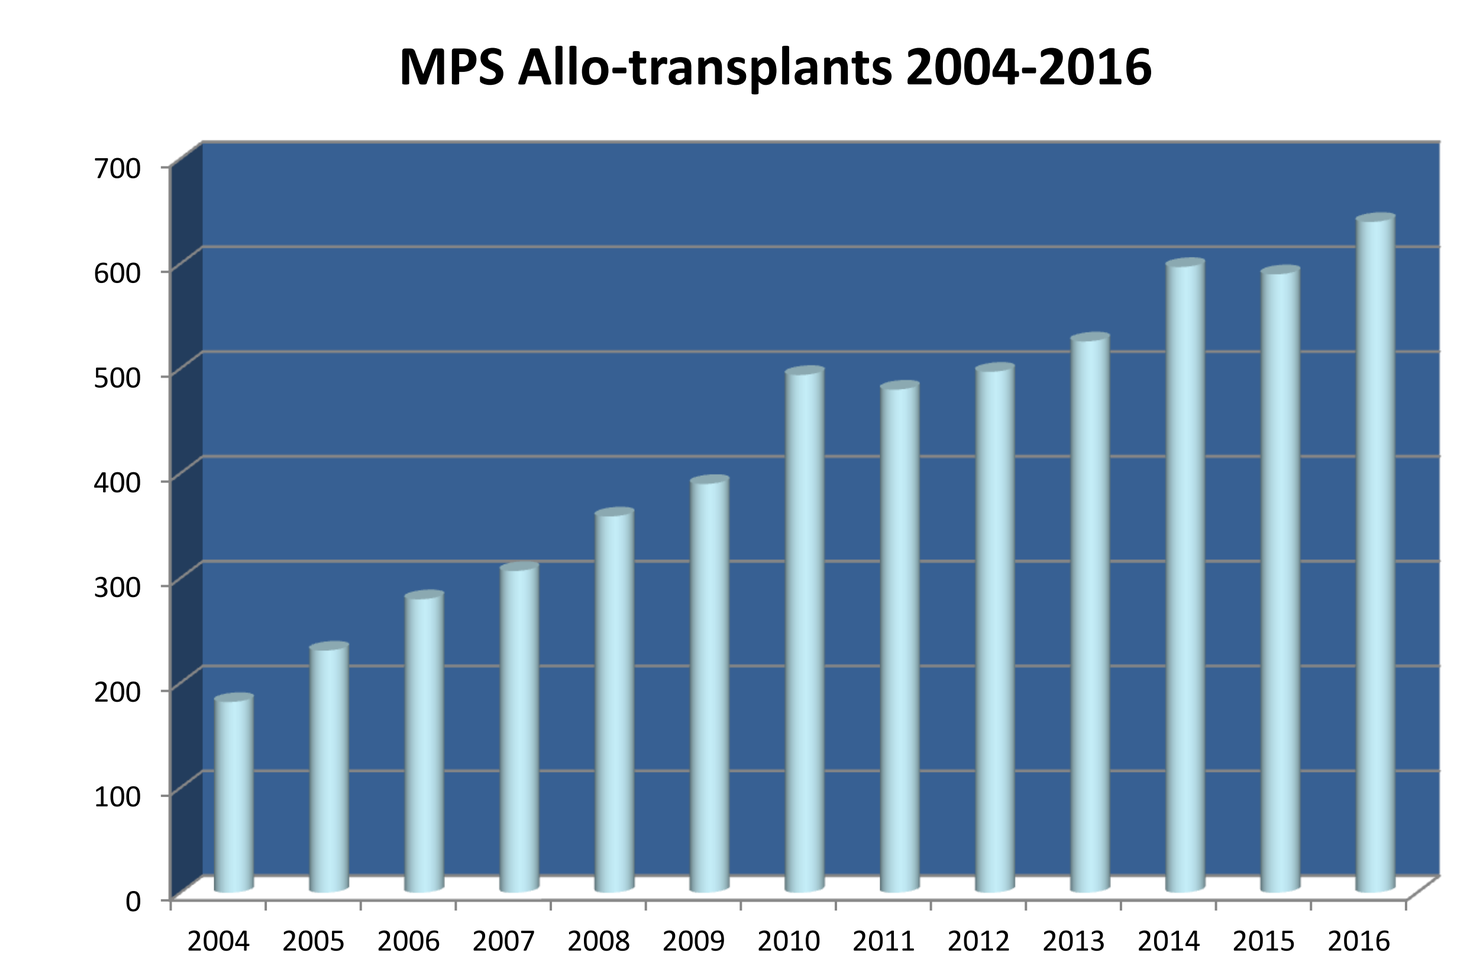
MPS Allo-transplants 2004-2016
0
100
200
300
400
500
600
700
2004 2005 2006 2007 2008 2009 2010 2011 2012 2013 2014 2015 2016

MPN Data Quality Initiative

This DQI focusses on the impact of pre-graft treatment on the response of the post-graft outcome. Essential data i.e. pre-treatment, cytogenetics, comorbidities, GvHD prophylaxis, as well as follow up information have been completed for 400 patients allowing to do 3 studies

Transplant indication for CML in 2018: Y. Chalandon
CMWP working party session.
Monday, March 19th, 09.50-10.10 Room: 3A

AlloHSCT in patients with CML-CP in the era of 3rd generation TKI: a study by the CMWP of the EBMT. Y. Chalandon et al.
MDS/MPN Session.
Monday March 19th, 17.20-17.30 Room: 5B

The data suggest that the number of TKI given prior to alloHSCT has no impact on post-transplant outcomes. Patients receiving 3rd Generation TKI might have worse outcomes. Patients in CP1 have better survival than more advanced CML patients. The performance status at transplant remains as an important predictive factor in the era of 3rd generation TKI.

Transplant-related prediction of survival in primary myelofibrosis: a study by the CMWP of the EBMT. N. Gagelmann et al.
MDS/MPN Session.
Monday March 19th, 17.30-17.40 Room: 5B

By using EBMT registry data it shows that available scores provide moderate prognostic ability to predict outcome of PMF patients undergoing alloHSCT. Furthermore, 8 factors that showed impact on OS were identified, which will be integrated in a patient- and transplant-related score in further analyses.

Outcome of MAC and RIC alloHSCT in myelofibrosis: a study by the CMWP of the EBMT. D. McLornan et al.
MDS/MPN Session.
Monday March 19th, 17.40-17.50 Room: 5B

This EBMT registry study of allo-HSCT for MF is the largest cohort reported to date. Historically, the impact of conditioning dose intensity on outcome in MF patients has been unclear. We observed no statistically significant differences between engraftment, GVHD rates, NRM, PFS and OS between these two large RIC and MAC cohorts .

In vivo T-cell depletion in patients with myelofibrosis transplanted from an HLA-matched sibling donor: a study by the CMWP of the EBMT. M. Robin et al.
MDS/MPN Session.
Monday March 19th, 18.00-18.10 Room: 5B

In myelofibrosis patients, TCD prevent acute GVHD without decreasing incidence of grade III-IV acute GVHD and chronic GVHD while it increases the relapse risk. Survivals are not significantly different without TCD suggesting that TCD does not improve outcome of myelofibrosis patients transplanted from an HLA matched sibling donor.

Recently published

Outcome after allo-SCT for Ph negative CML.
F. Onida. et al. *BJH* 2017;177:759-765

Update of T315I mutation (vs Ponatinib).
G. Basak / F Nicolini et al.. Cancer 2017;123:2875-2880.

#EBMT18
9
www.ebmt.org