European Society for Blood and Marrow Transplantation

CALM
Geographical Overview of Follow up Data Collection

CALM GeoData Overview Annual FU 2008 - 2015

In total 48 hospitals from 19 different countries are participating in the CALM Study, resulting in 7802 patients . Currently we are in the final stages of the data collection process. The data submission deadline is **April 2016**. After cleaning and checking the submitted data, the final database will be transferred to Sanofi in June 2016.

*** Please send in your data***

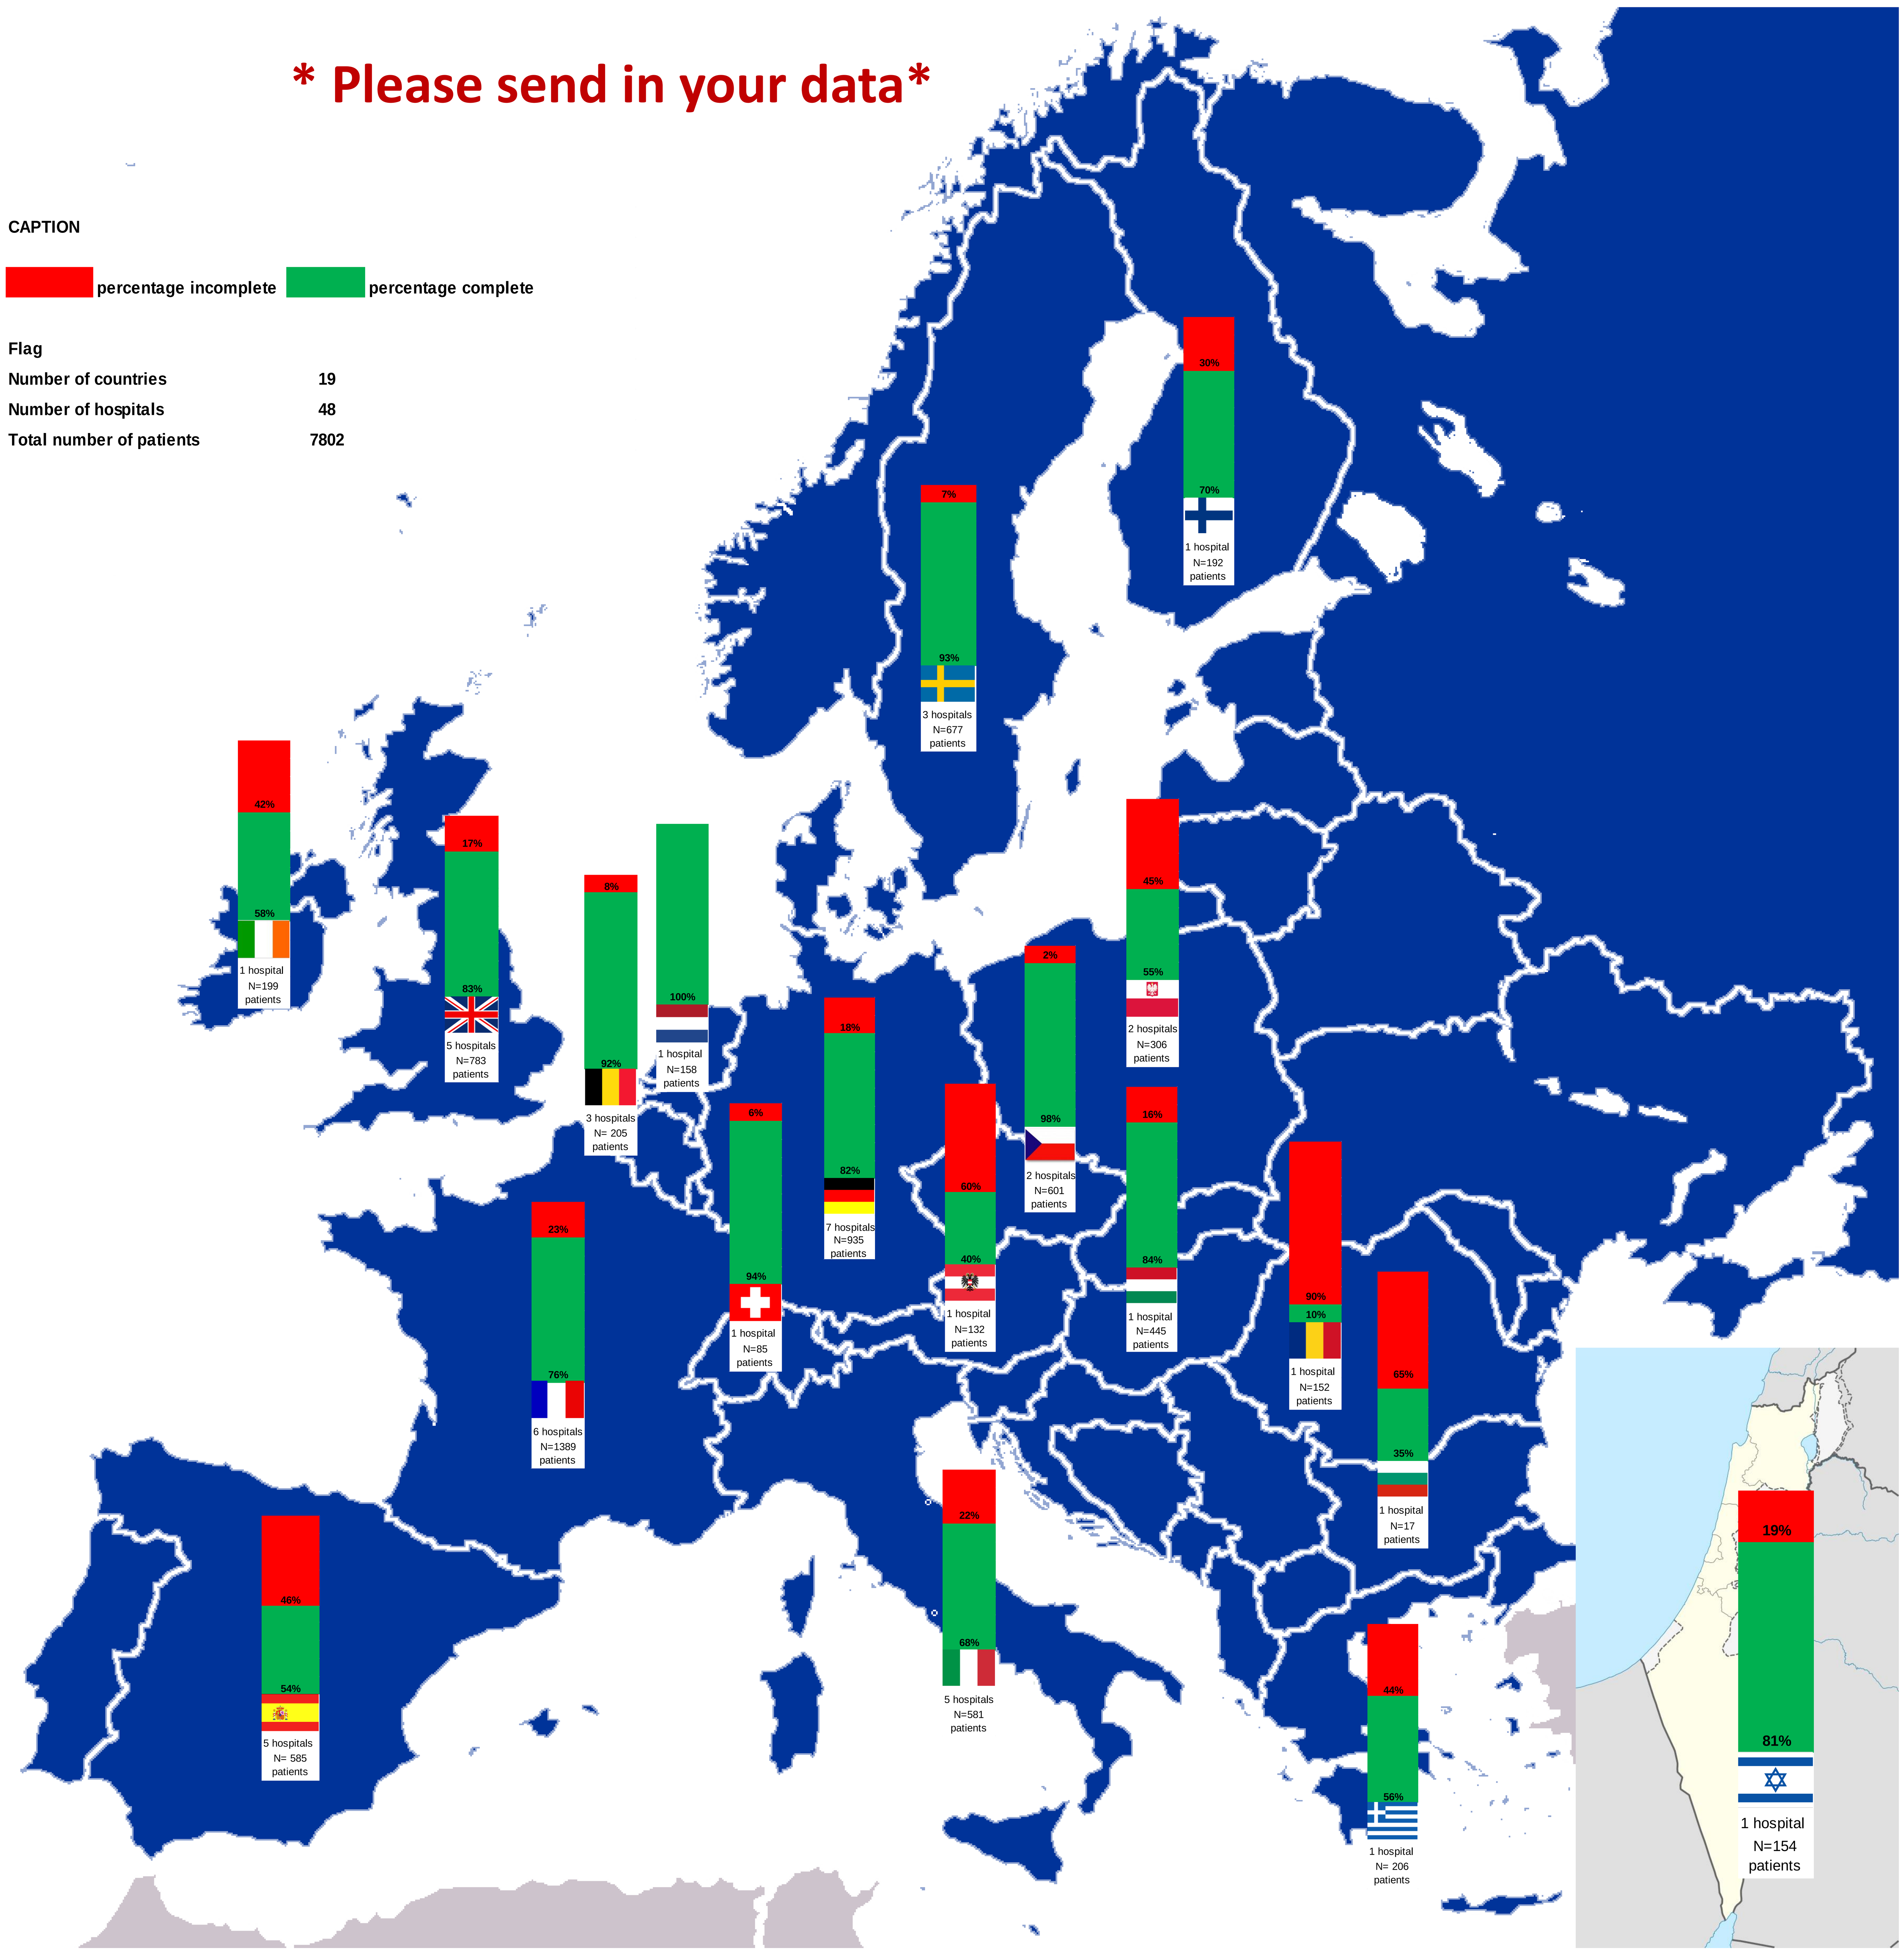

CAPTION
percentage incomplete
percentage complete
Flag
Number of countries 19
Number of hospitals 48
Total number of patients 7802
42% 58% 1 hospital N=199 patients
17% 83% 5 hospitals N=783 patients
8% 92% 3 hospitals N= 205 patients
100% 1 hospital N=158 patients
23% 76% 6 hospitals N=1389 patients
46% 54% 5 hospitals N= 585 patients
6% 94% 1 hospital N=85 patients
18% 82% 7 hospitals N=935 patients
7% 93% 3 hospitals N=677 patients
30% 70% 1 hospital N=192 patients
60% 40% 1 hospital N=132 patients
2% 98% 2 hospitals N=601 patients
45% 55% 2 hospitals N=306 patients
16% 84% 1 hospital N=445 patients
90% 10% 1 hospital N=152 patients
65% 35% 1 hospital N=17 patients
22% 68% 5 hospitals N=581 patients
44% 56% 1 hospital N= 206 patients
19% 81% 1 hospital N=154 patients

EBMT studies based on CALM data

With the CALM data the EBMT Lymphoma and Chronic Malignancies Working Parties are also planning/perfoming other scientific studies.

Lymphoma WP

- Relapse after auto SCT for DLBCL
- AutoSCT in HIV in the cART era
- FL relapse after autoSCT
- BV as bridge to transplant in ALCL
- High dose Chemotherapy and autoSCT in nodular lymphocyt-predominant Hodgkin Lymphoma

Chronic Malignancies WP

- Influence of induction on OS
- Mobilization: G-CSF alone vs G-CSF + chemo
- Conditioning: NMA vs RIC vs MAC
- Single vs Double auto SCT
- Allogeneic transplant after autologous transplant
- Influence of Cytogenetic abnormalities in MM
- Renal impairment & Karnofsky score & Comorbidities
- Secondary malignancies and second diagnosis after first having MM
- Investigate IG's in CALM
- Older patients in the CALM group

calmebmt@lumc.nl

#EBMT16

www.ebmt.org