Lymphoma Working Party

General activity

Members and staff: Peter Dreger (Chair), Stephen Robinson (Secretary), Olivier Hermine, Silvia Montoto, Norbert Schmitz, Harry Schouten, Anna Sureda, Alina Tanase, Ariane Boumendil (Statistician), Hervé Finel (Study Coordinator)

The EBMT Database 2015: Transplants by disease category

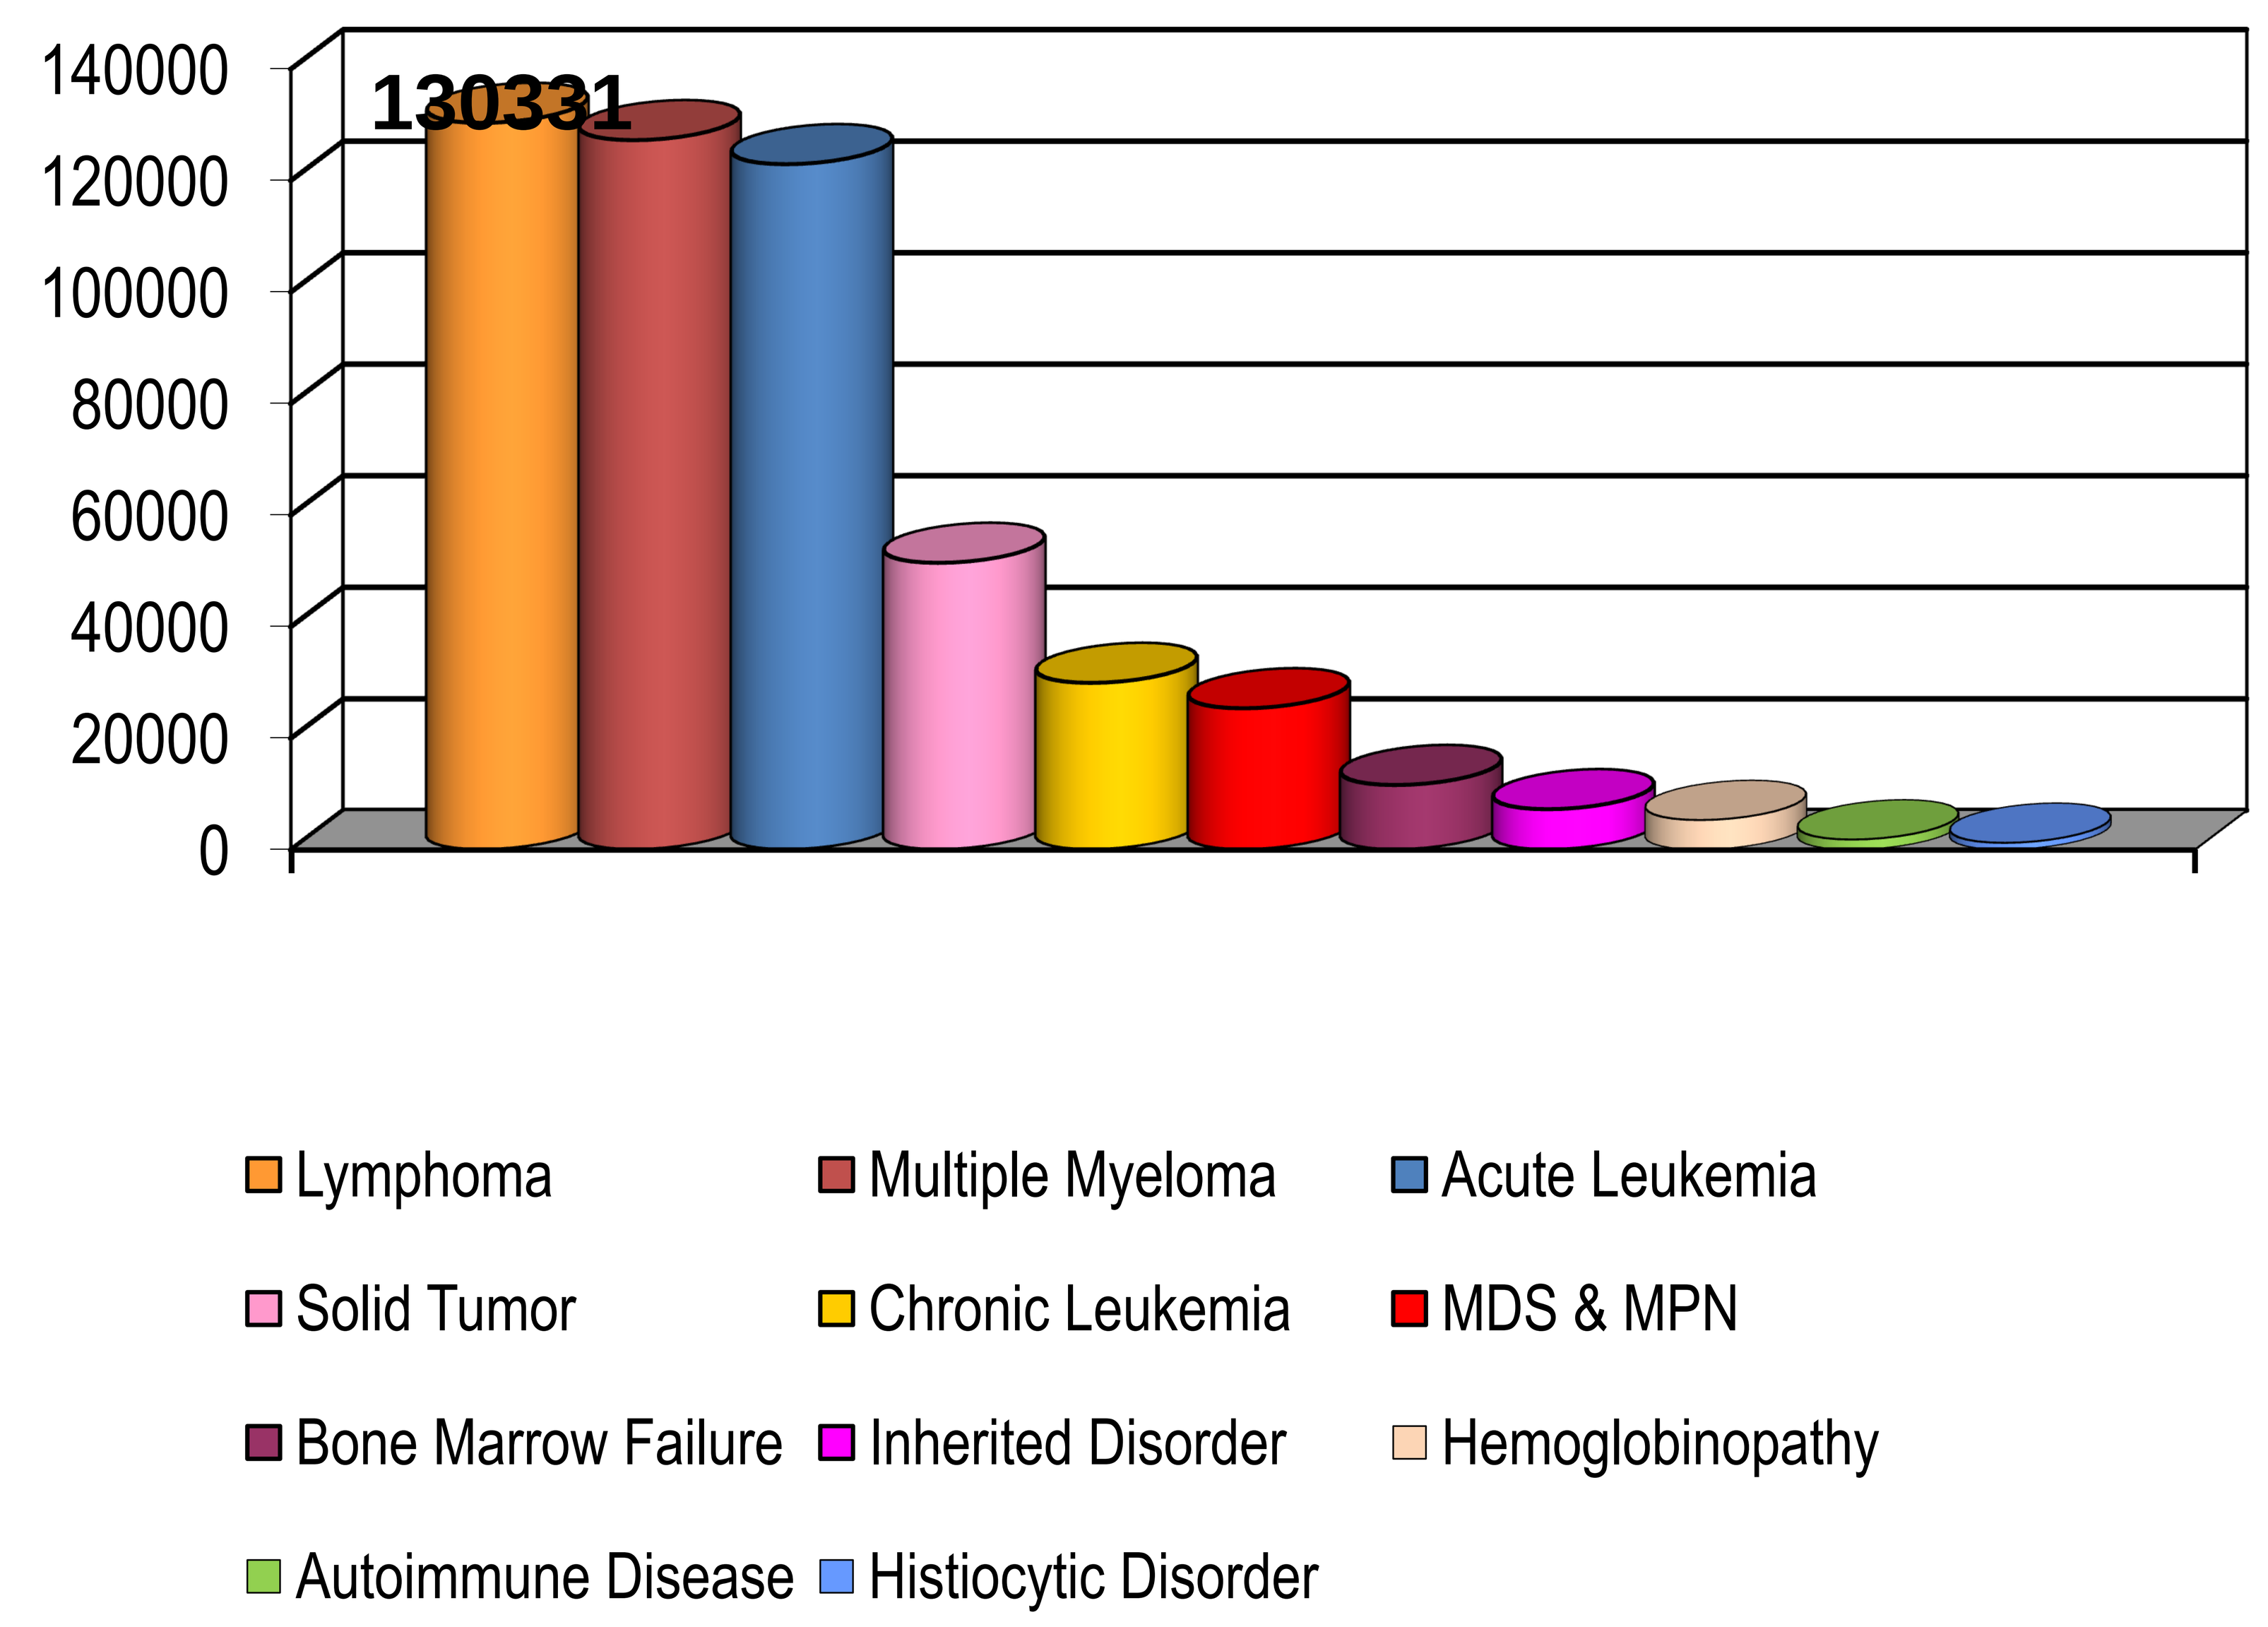

130331
140000
120000
100000
80000
60000
40000
20000
0
Lymphoma
Multiple Myeloma
Acute Leukemia
Solid Tumor
Chronic Leukemia
MDS & MPN
Bone Marrow Failure
Inherited Disorder
Hemoglobinopathy
Autoimmune Disease
Histiocytic Disorder

Transplant activity for lymphoma EBMT 2001-2012

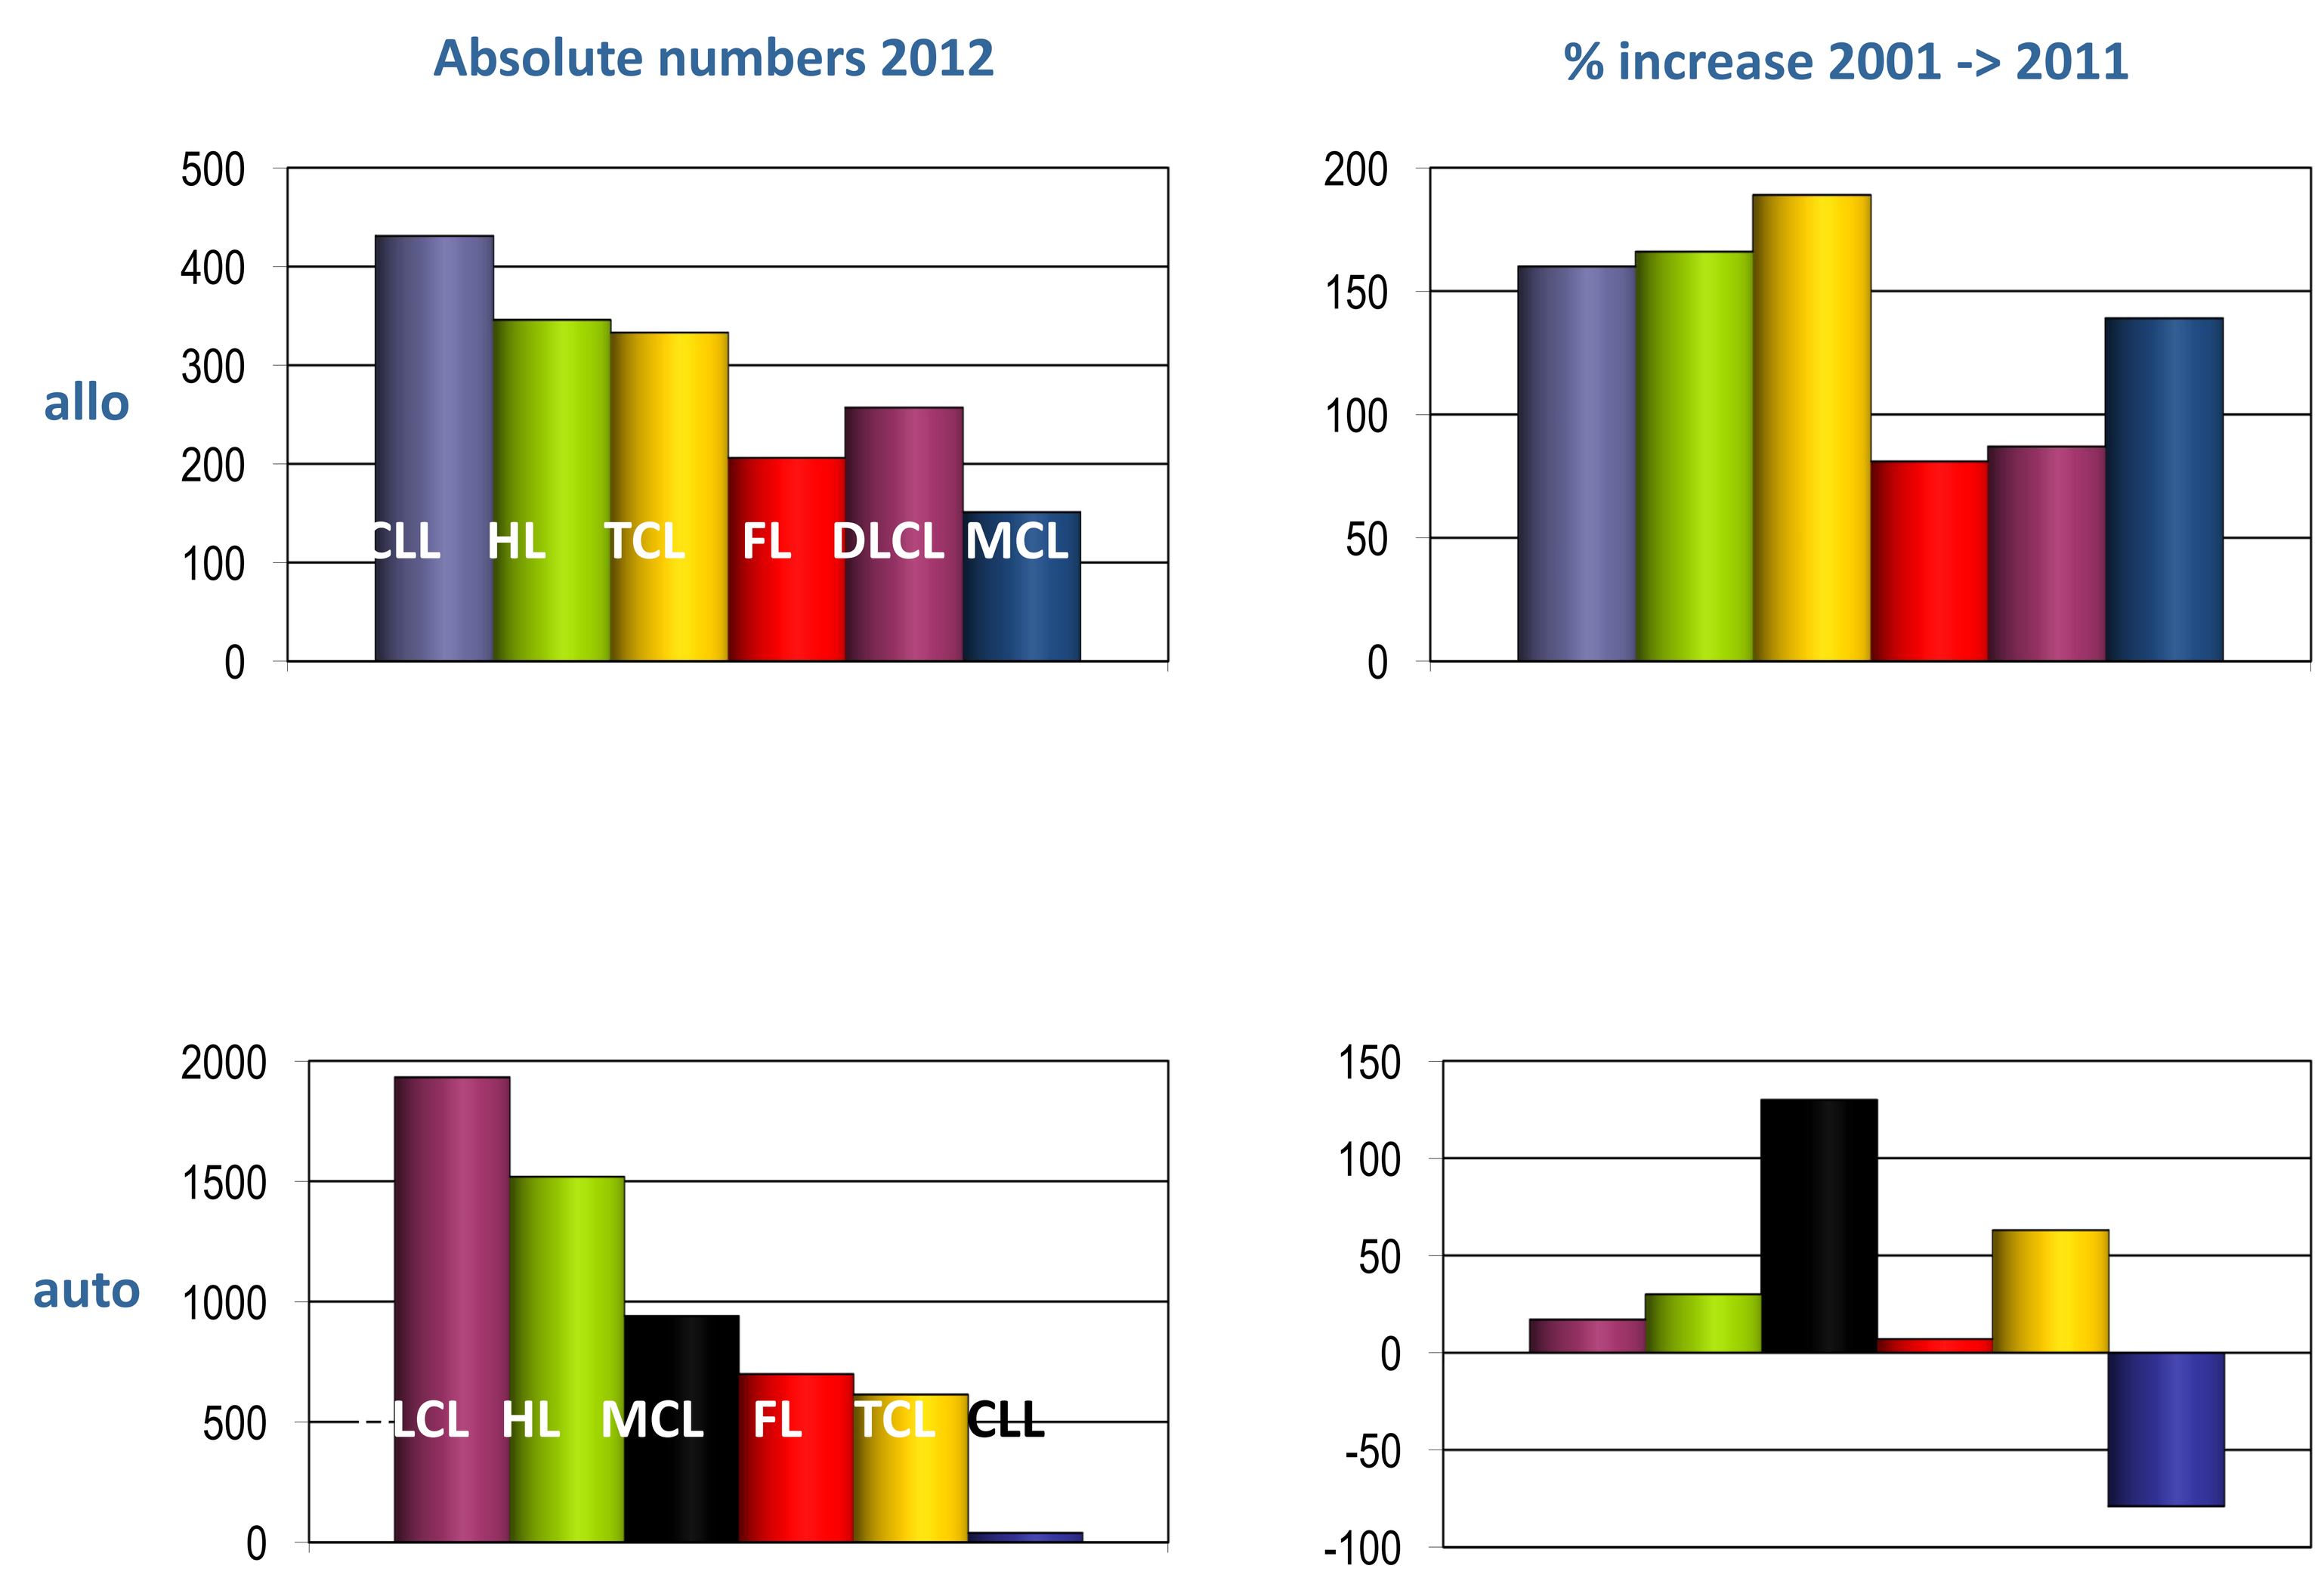

Absolute numbers 2012
% increase 2001 -> 2011
allo
CLL HL TCL FL DLCL MCL
auto
DLCL HL MCL FL TCL CLL

Total lymphoma transplants 2012 (w/o CLL): **allo 1427; auto 6177**

Conditioning Regimens for Lymphomas (HL, DLBCL, FL, MCL) in the ASCT Setting

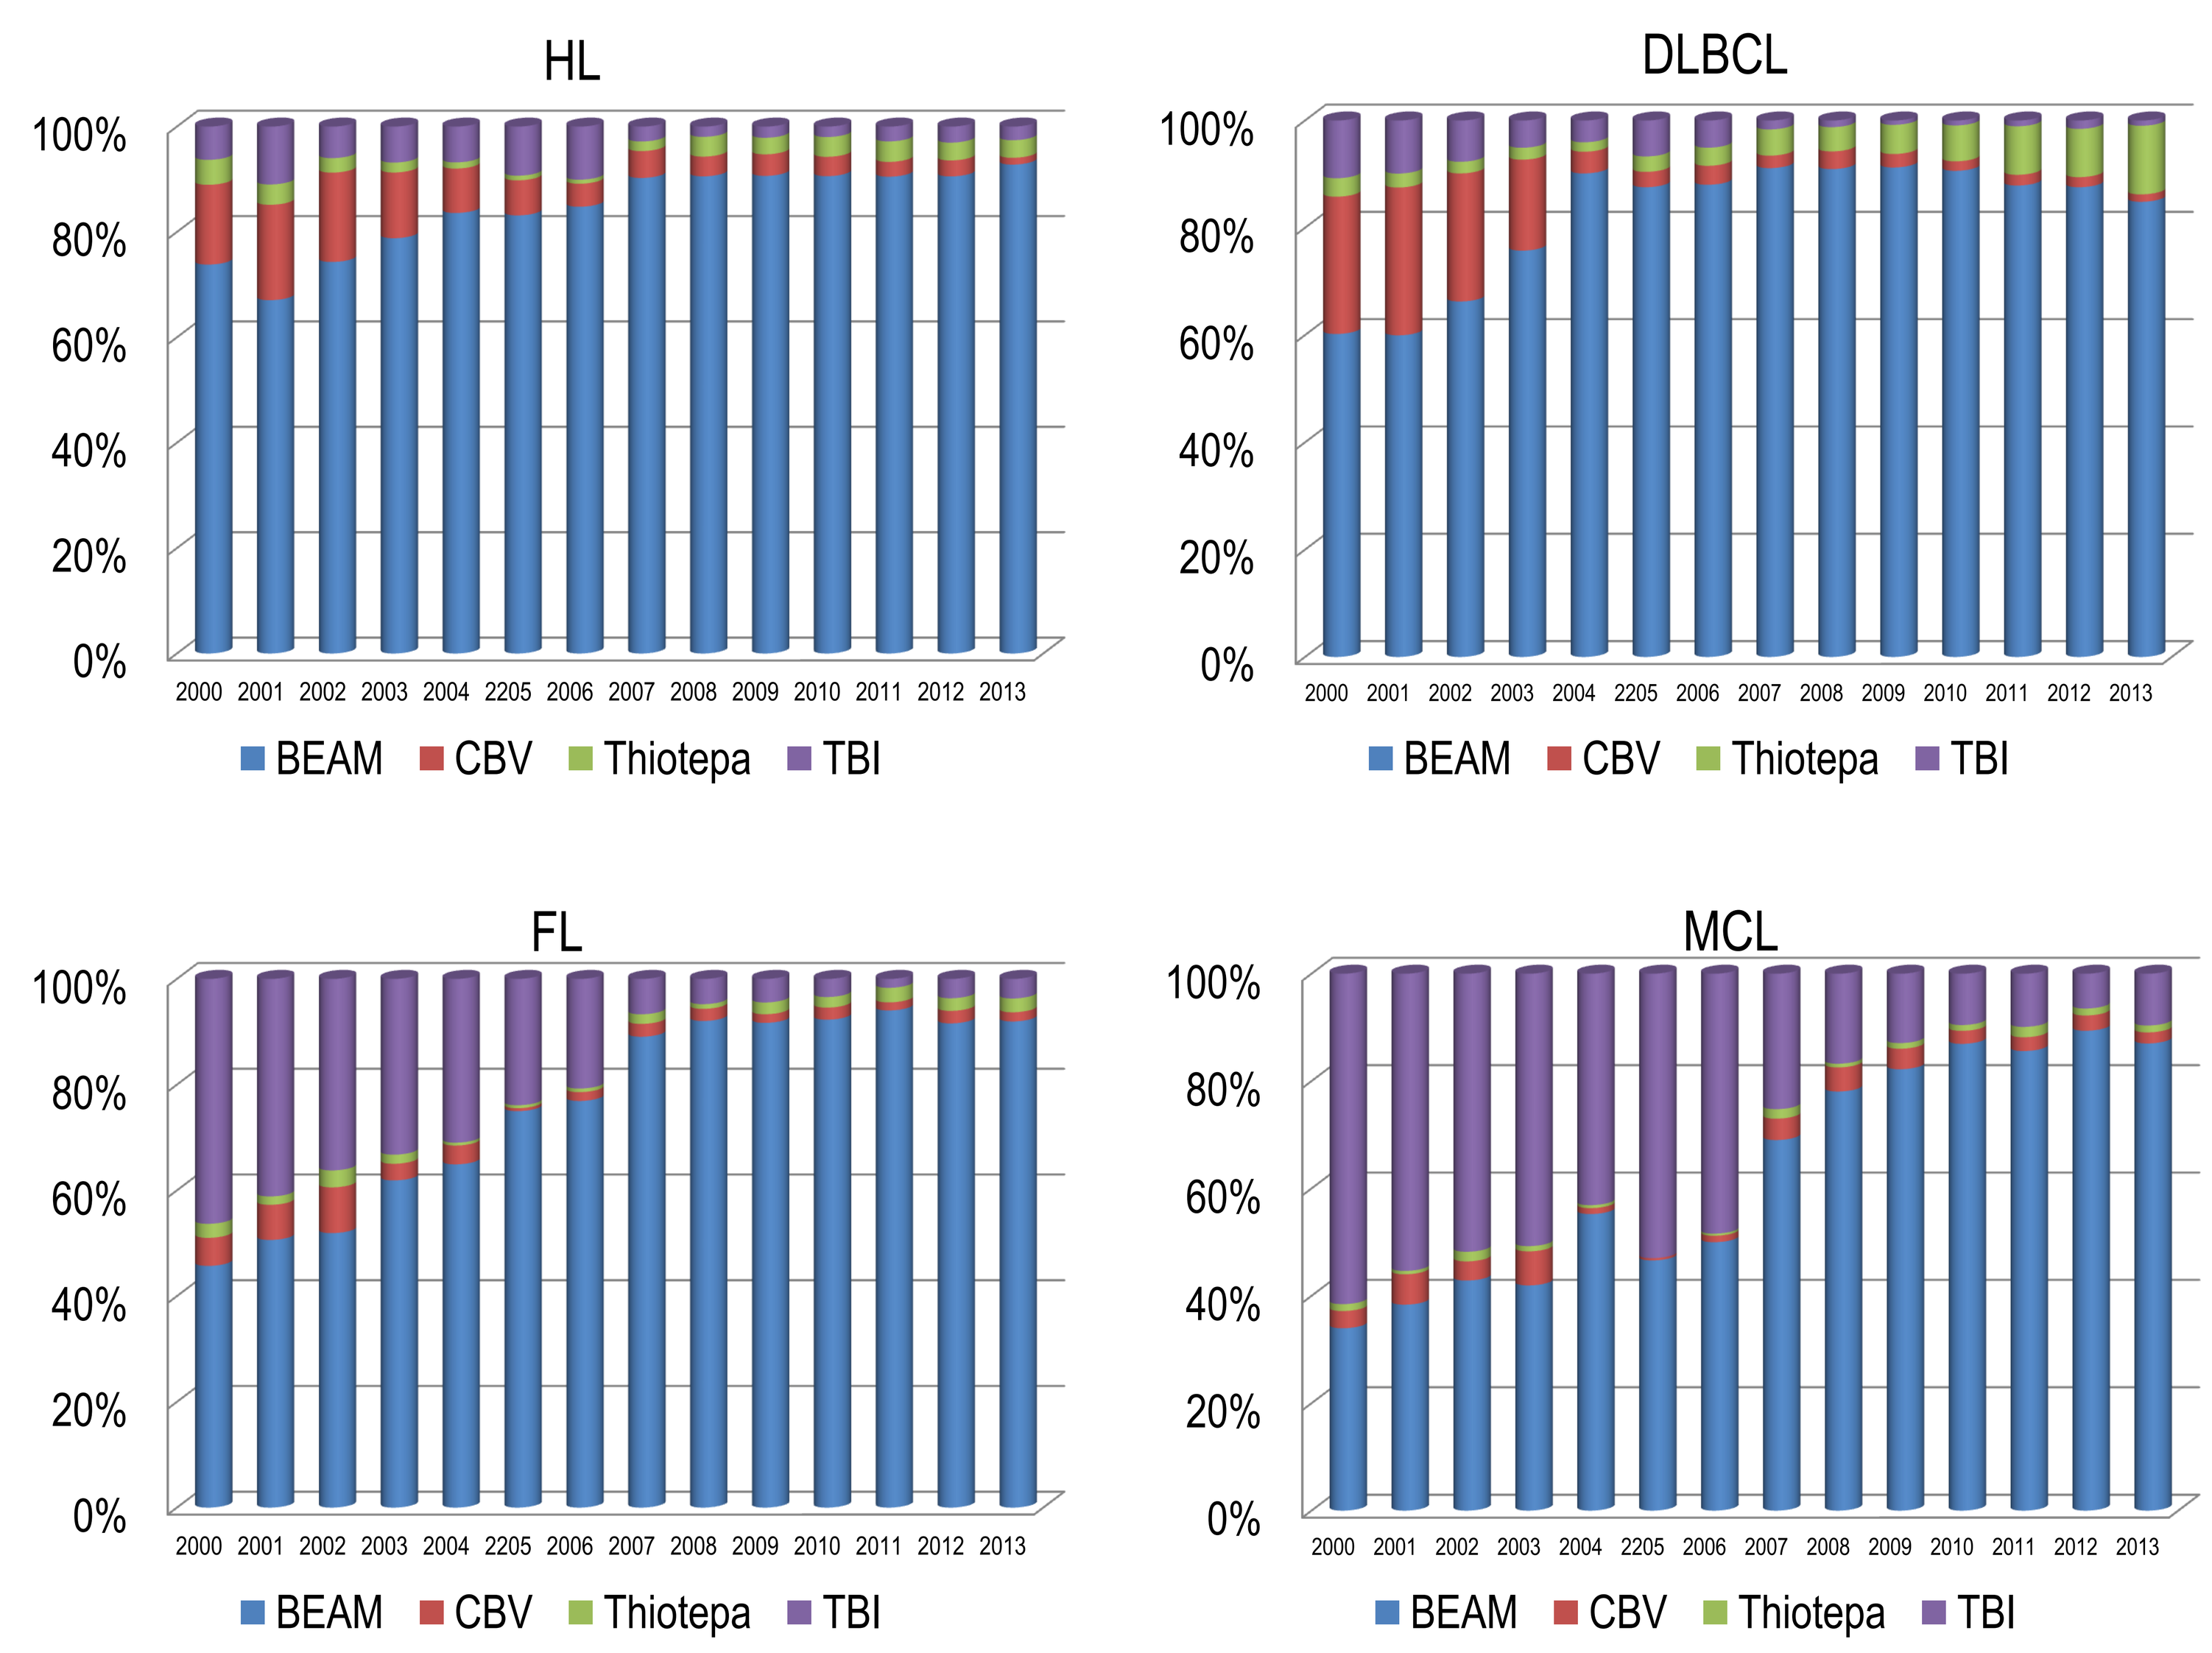

HL
DLBCL
FL
MCL
2000 2001 2002 2003 2004 2205 2006 2007 2008 2009 2010 2011 2012 2013
BEAM
CBV
Thiotepa
TBI

Conditioning Regimens for Lymphomas (HL, DLBCL, FL, MCL) in the RIC-Allo Setting

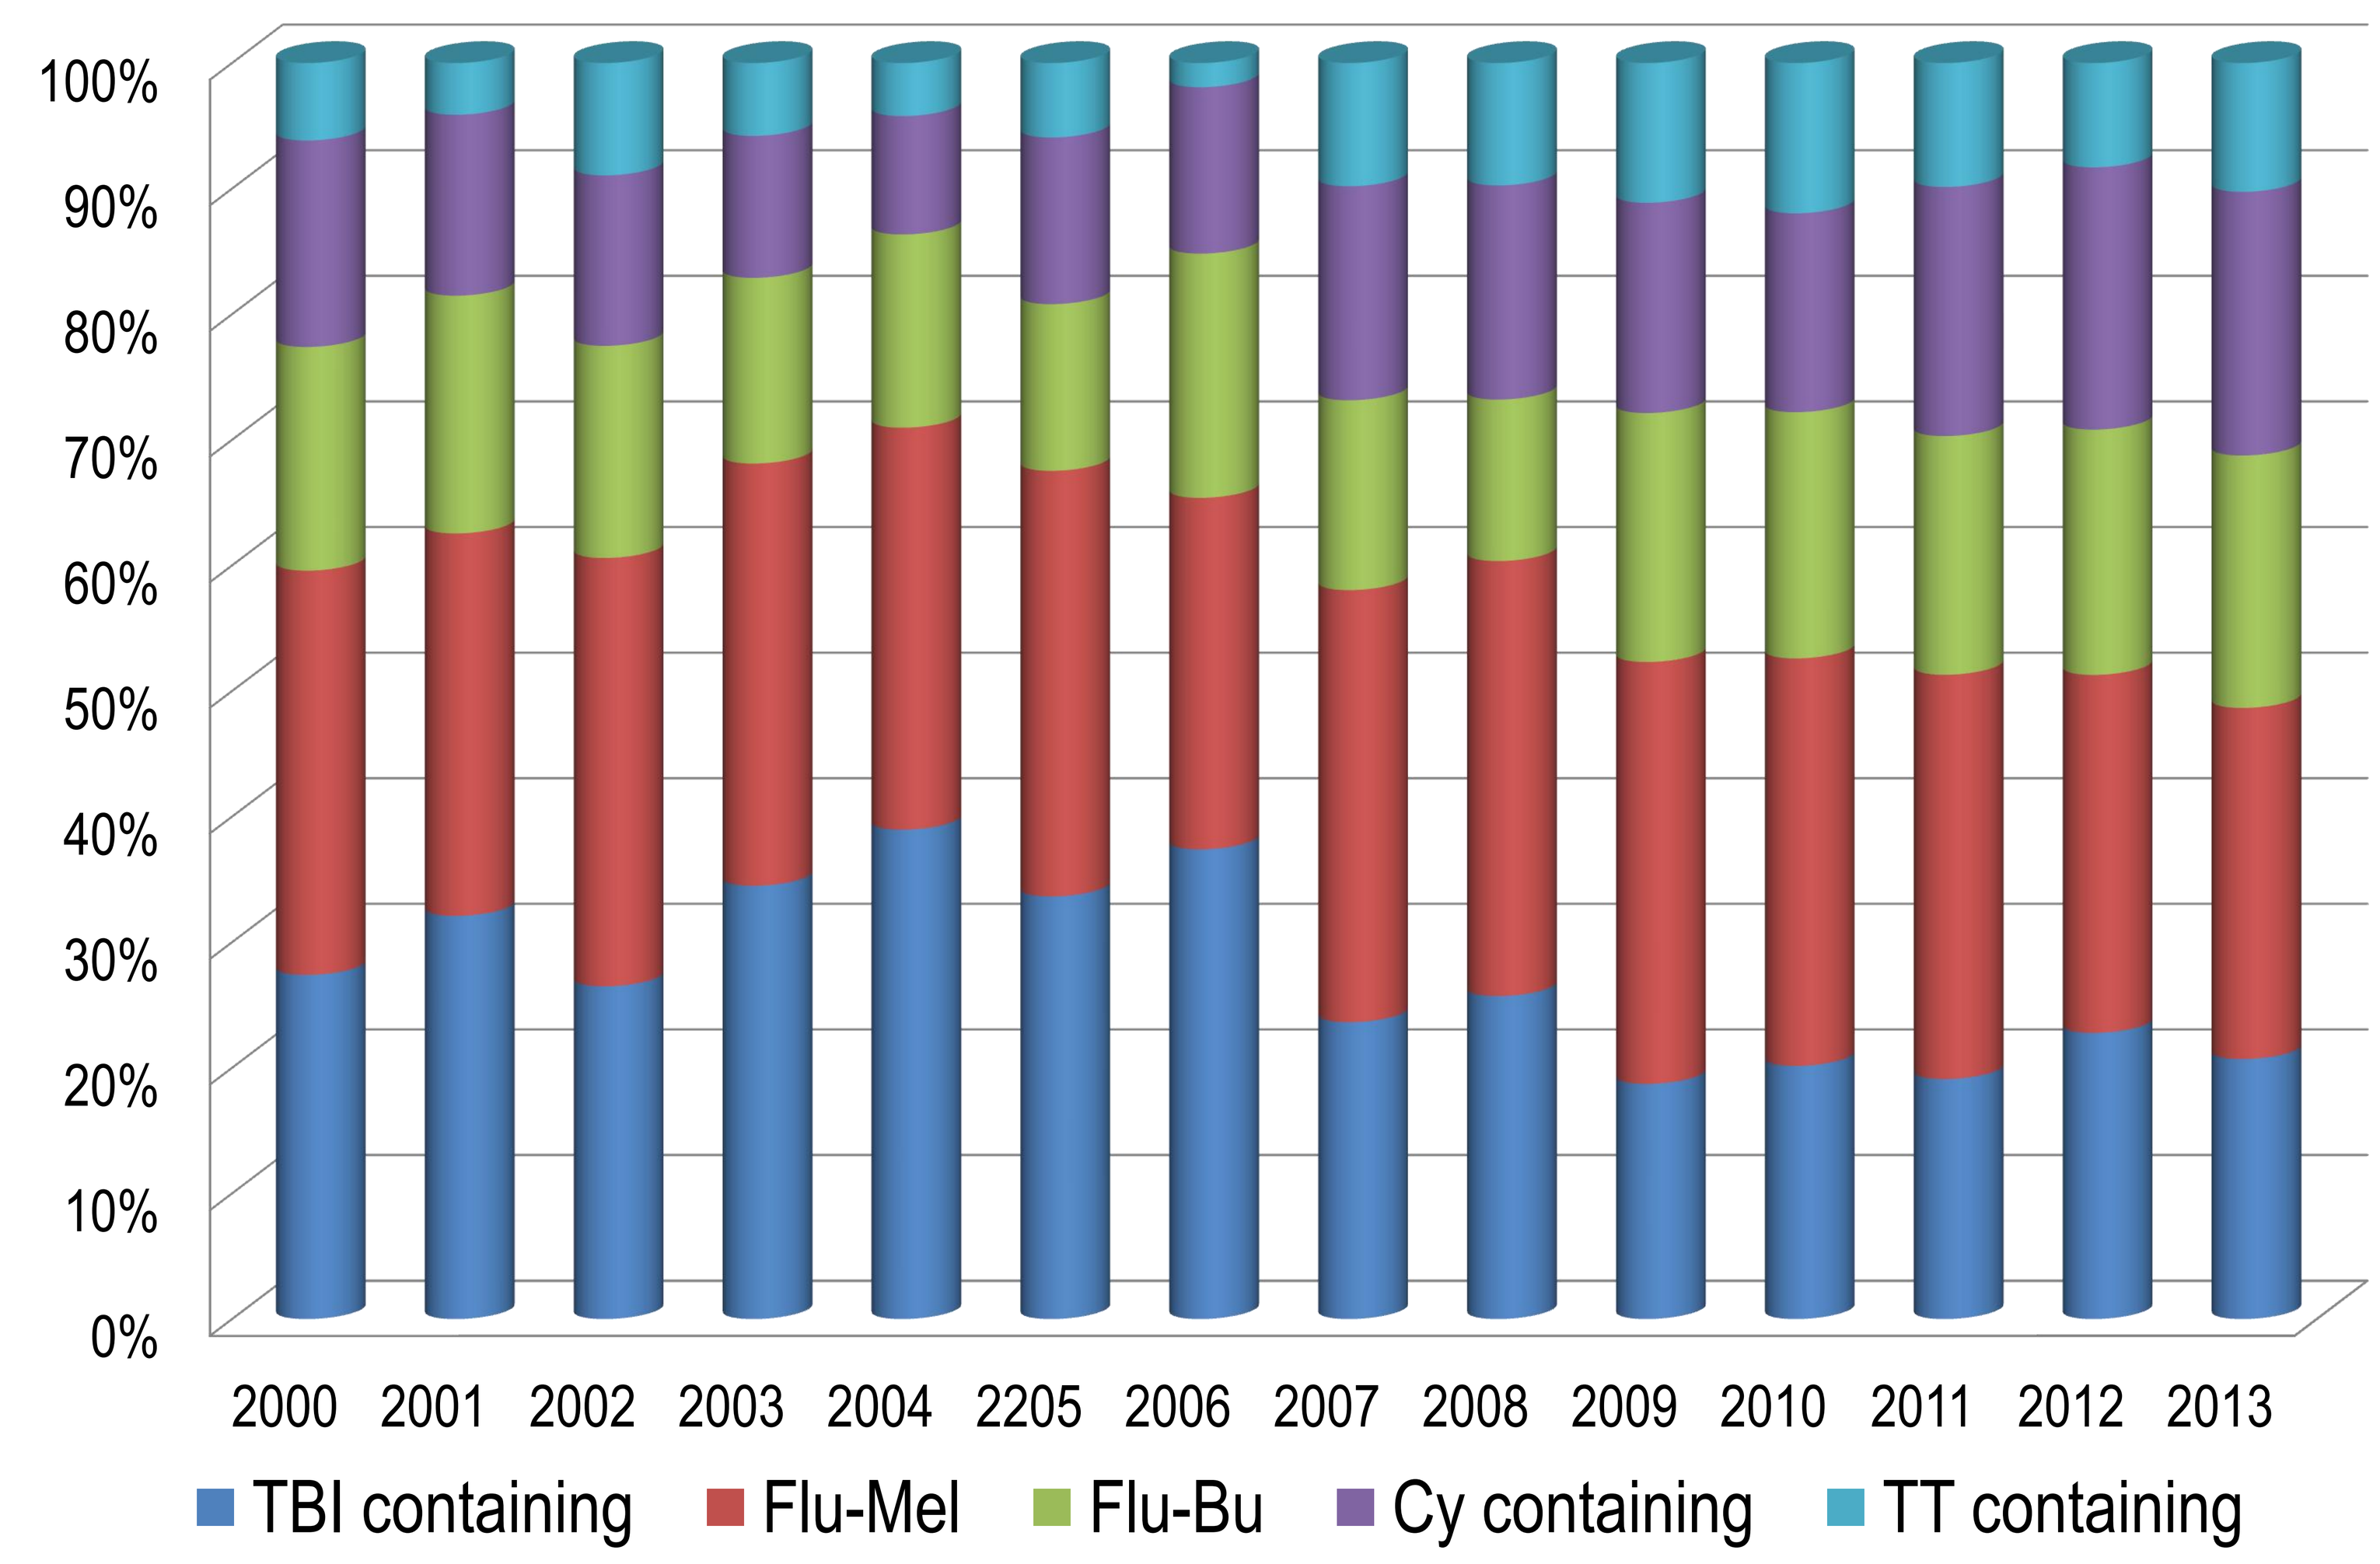

2000 2001 2002 2003 2004 2205 2006 2007 2008 2009 2010 2011 2012 2013
TBI containing
Flu-Mel
Flu-Bu
Cy containing
TT containing

Lymphoma Registry: Allo-SCT adults 18-50 yo

NRM (%) by period according to donor type

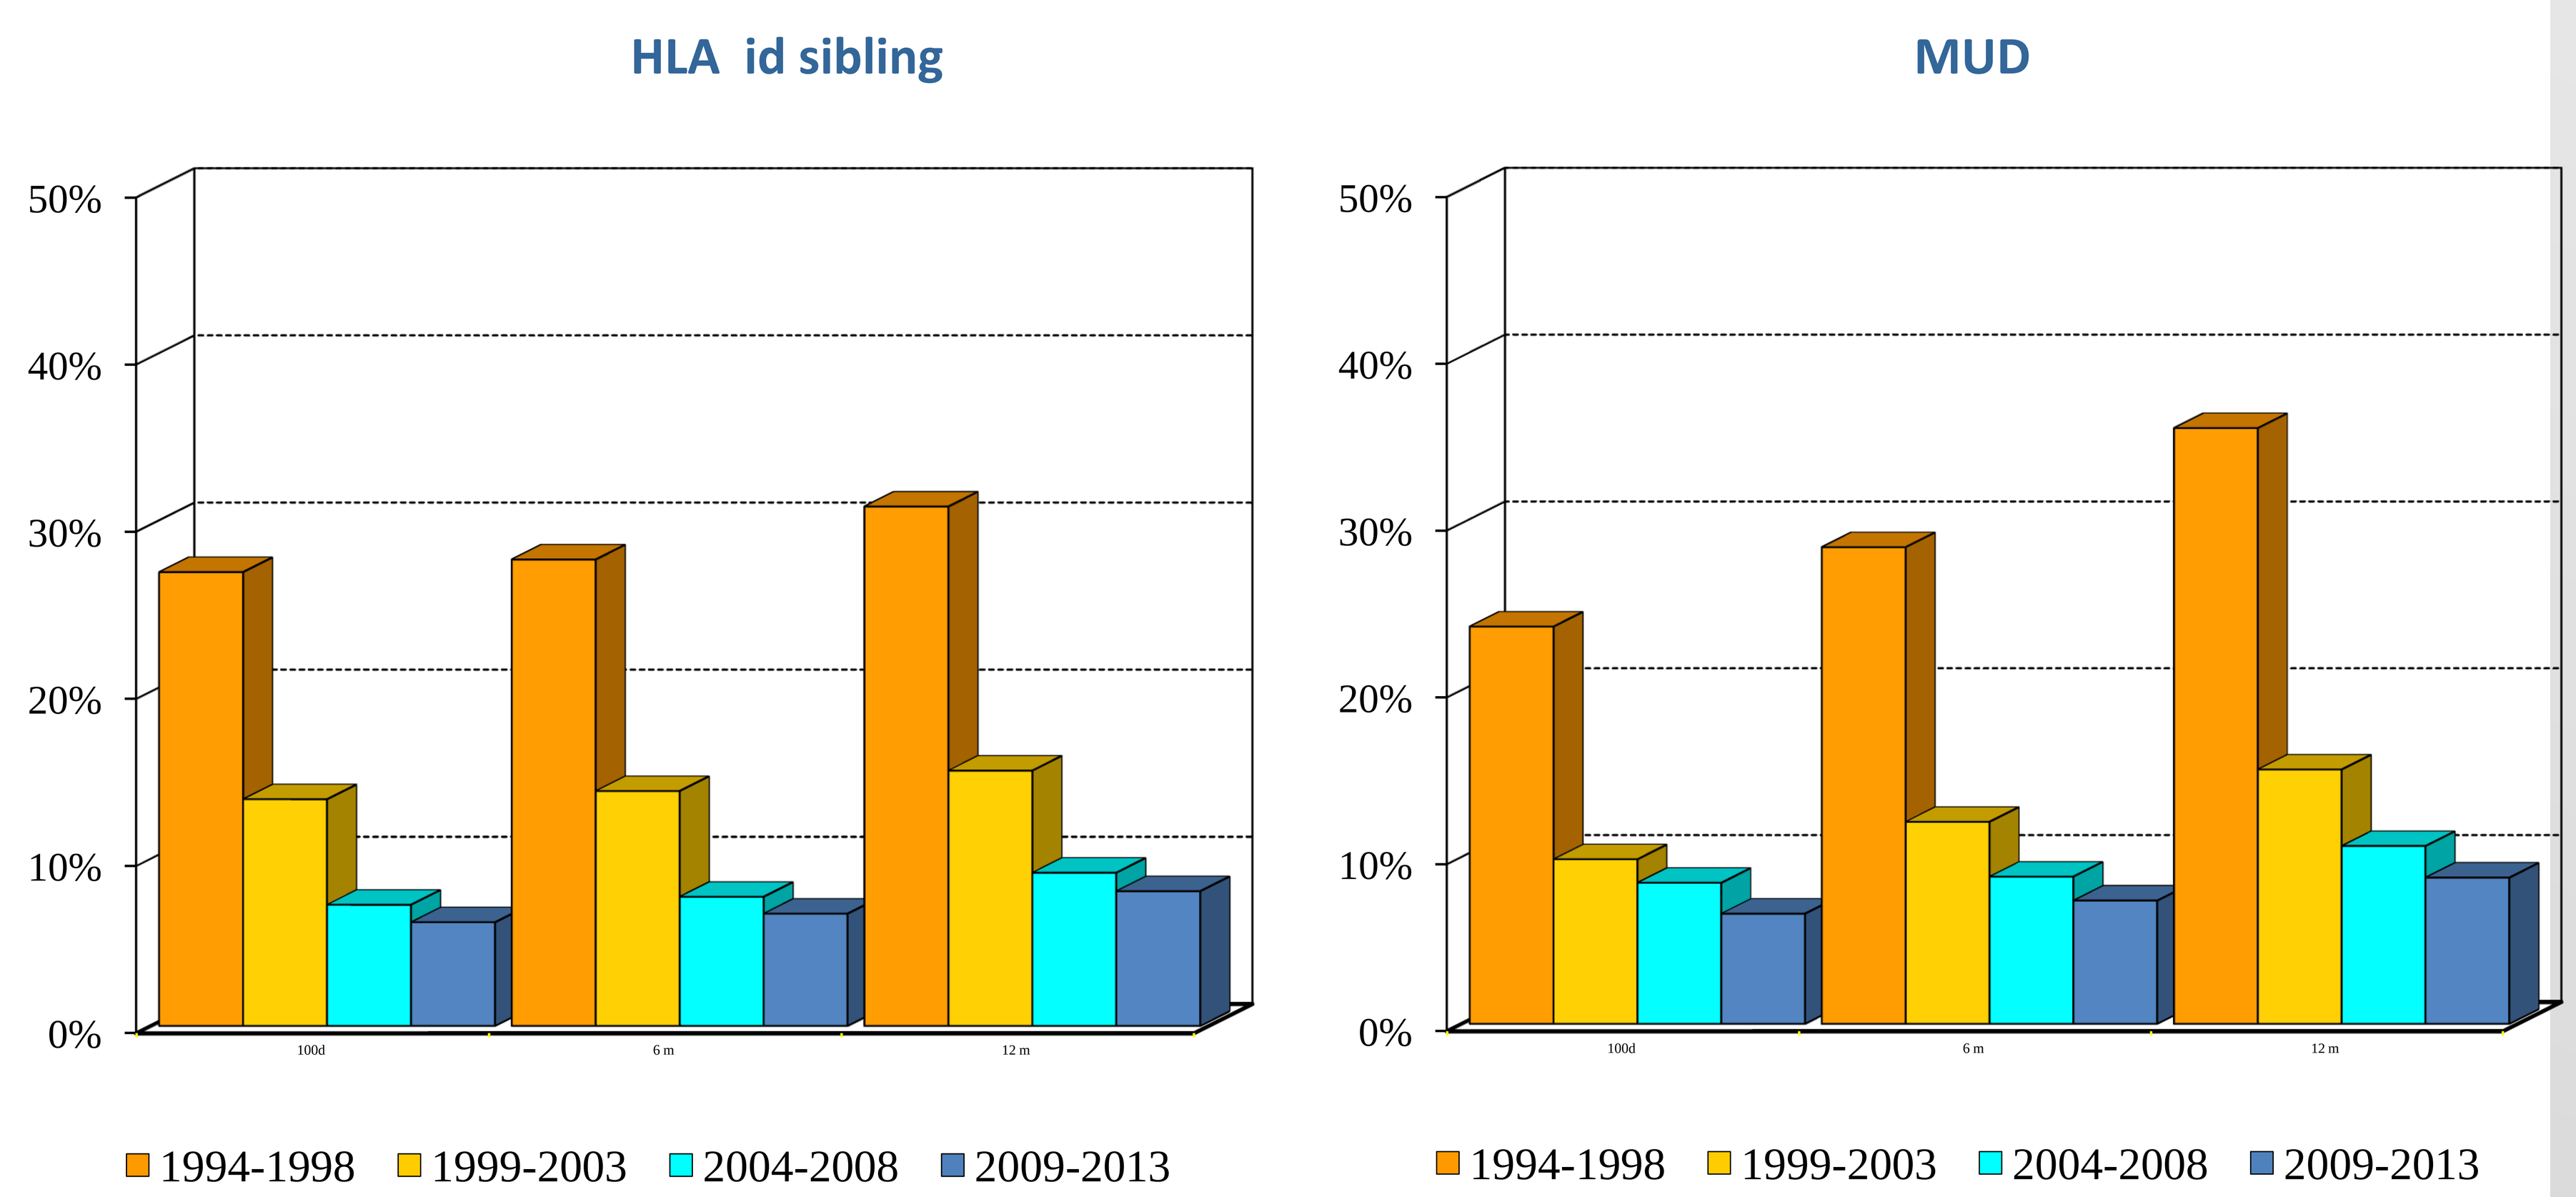

HLA id sibling
MUD
1994-1998
1999-2003
2004-2008
2009-2013

Lymphoma Registry: Allo-SCT adults 51-70 yo

NRM (%) by period according to the intensity of conditioning regimen

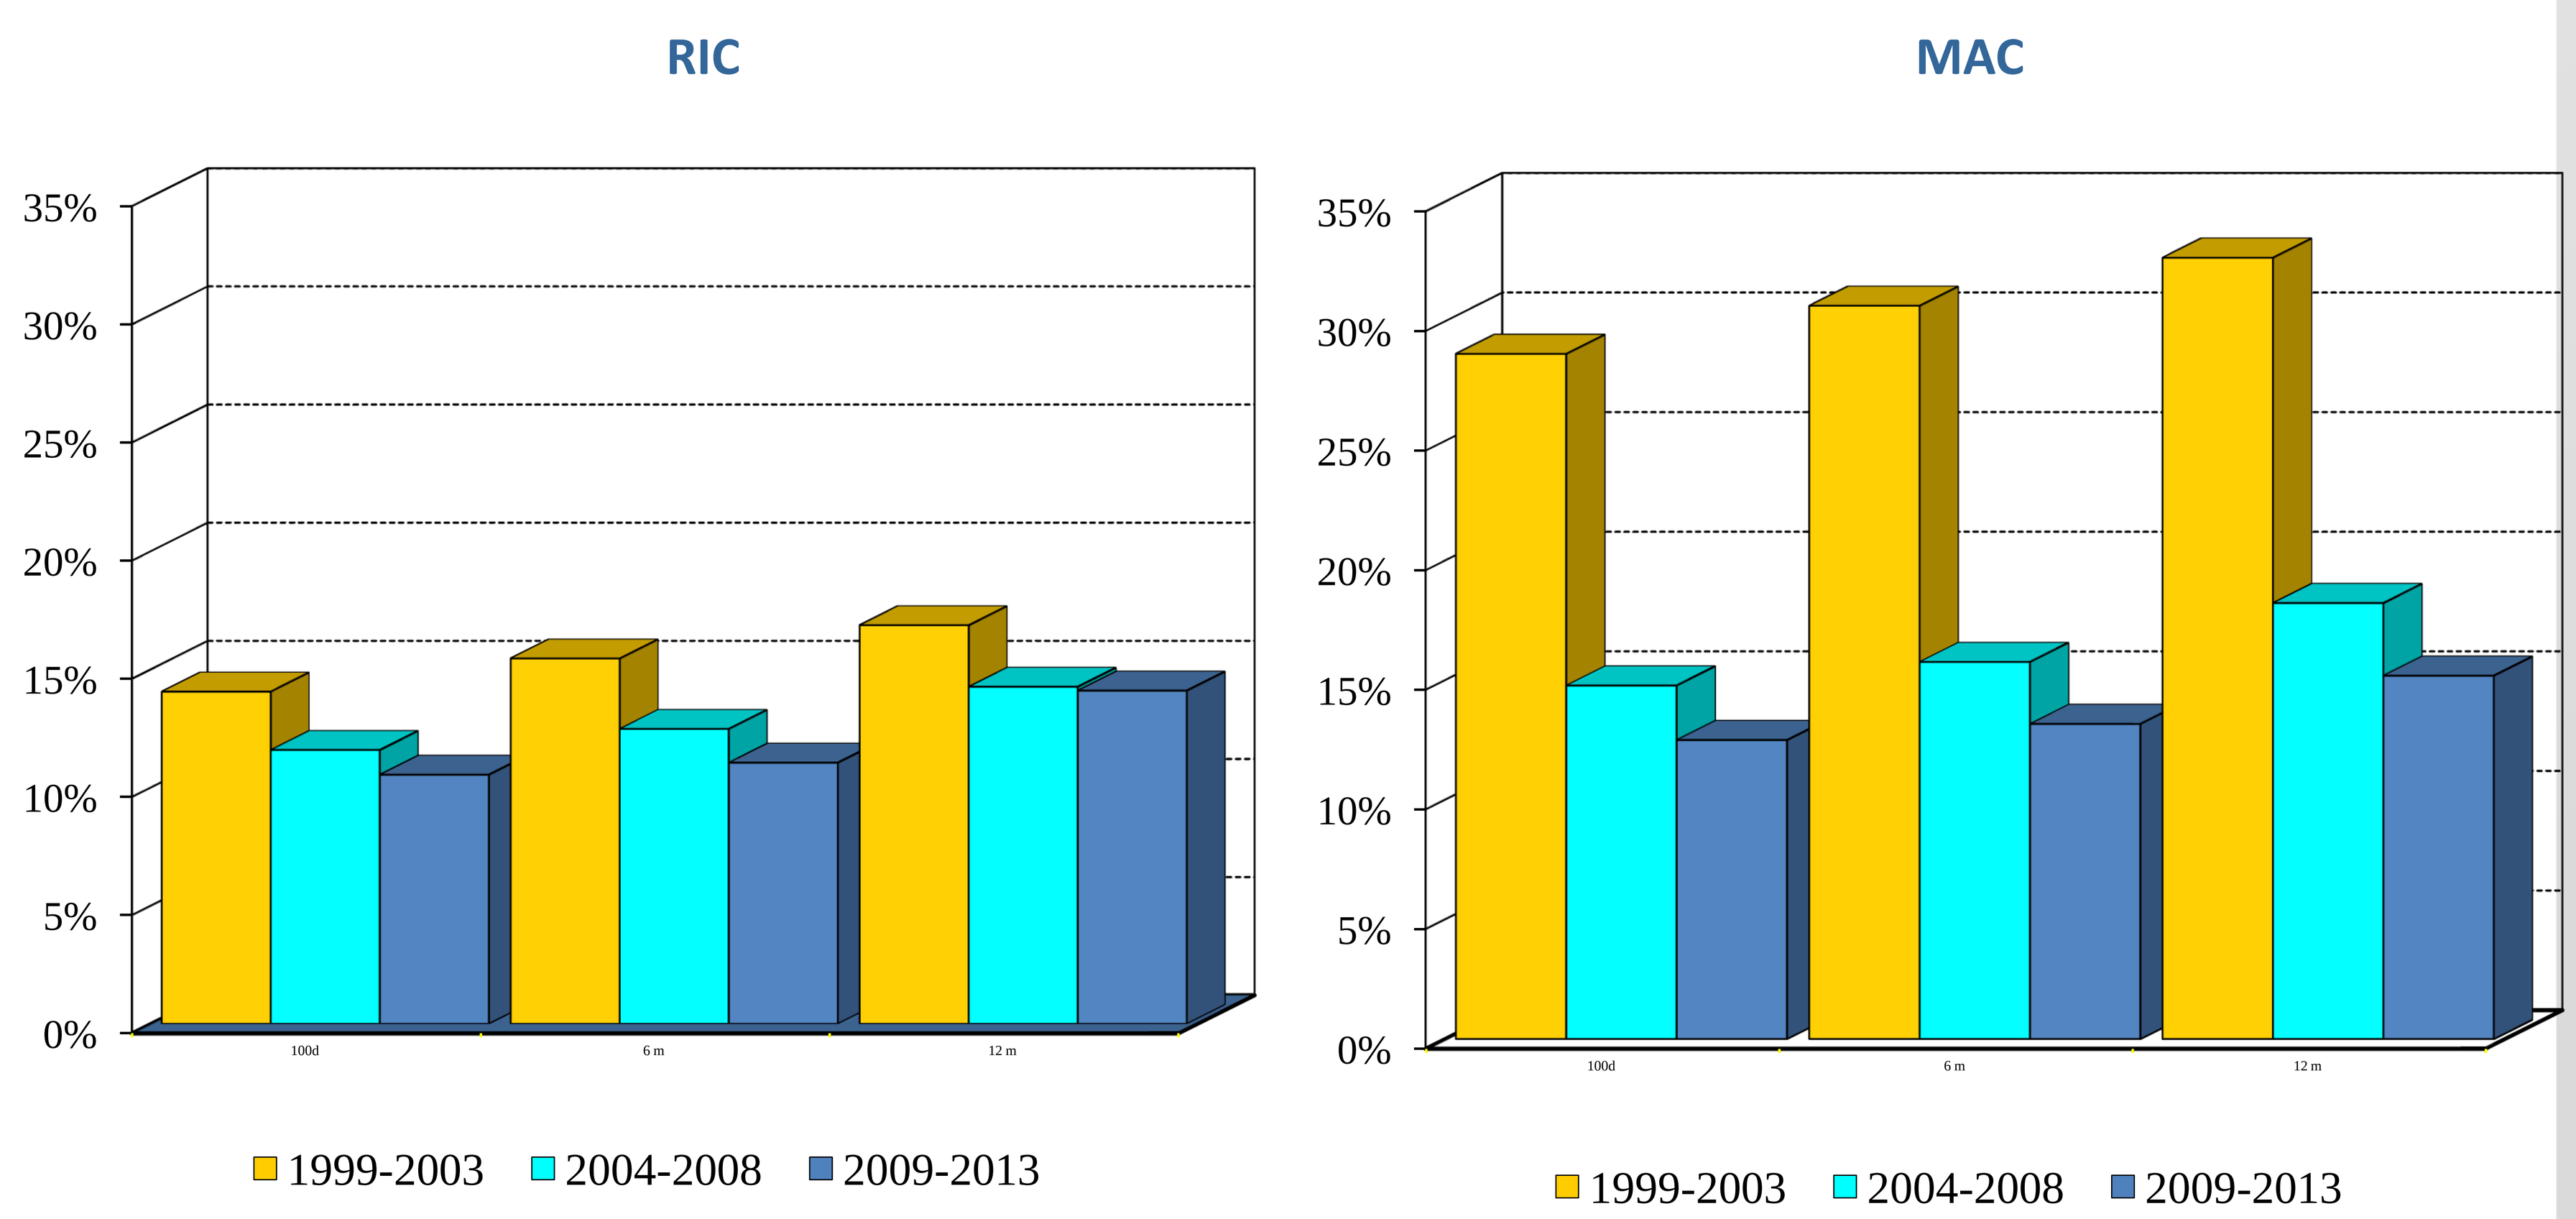

RIC
MAC
1999-2003
2004-2008
2009-2013

#EBMT15

www.ebmt.org