EBMT
European Society
for Blood and Marrow Transplantation

Inborn Errors Working Party

Chair: Arjan Lankester
Secretary: Michael Albert

Studies

Topic*	Disease Type	Joint
HSCT in LAD	PID	IEWP – PDWP
Outcome of Domino HSCT	PID	IEWP
HSCT in CGD	PID	IEWP
BuFlu vs. TreoFlu in WAS	PID	IEWP – Scetide
HSCT in IFNγ R1/R2-D	PID	IEWP – ESID – LASID – CIS ASID – PIDTC – Scetide
HSCT in porphyria	IMD	IEWP – CIBMTR
Outcomes after CBT in IMD	IMD	IEWP – Eurocord
HSCT in LAL-D (Wolman)	IMD	IEWP
Eculizumab for HSCT-TMA		PDWP – IEWP
HSCT in HLH adults	HLH	CMWP – IEWP
UCBT in children with FHLH	HLH	Eurocord – IEWP
Late effects after HSCT for HLH	HLH	PDWP – IEWP

** If you wish to participate in the data collection for these studies, please contact: IEWPebmt@lumc.nl*

Recent Publications on behalf of IEWP

Hematopoietic stem cell transplantation in patients with gain-of-function signal transducer and activator of transcription 1 mutations. *J Allergy Clin Immunol 2017*
J.W. Leiding*, S. Okada*, D. Hagin, M. Abinun, A. Shcherbina, D.N. Balashov, V.H.D. Kim, A. Ovadia, S.L. Guthery, M. Pulsipher, D. Lilic, L.A. Devlin, S. Christie, M. Depner, S. Fuchs, A. van Royen-Kerkhof, C. Lindemans, A. Petrovic, K.E. Sullivan, N. Bunin, S. Sebnem Kilic, F. Arpaci, O. de la Calle-Martin, L. Martinez-Martinez, J. C. Aldave, M. Kobayashi, T. Ohkawa, K. Imai, A. Iguchi, C. M. Roifman, A. R. Gennery, M. Slatter, H. D. Ochs, T. Morio**, and T. R. Torgerson**, on behalf of the Inborn Errors Working Party of the European Society for Blood and Marrow Transplantation and the Primary Immune Deficiency Treatment Consortium

Reticular dysgenesis: international survey on clinical presentation, transplantation, and outcome. *Blood 2017*
M. Hoenig, C. Lagresle-Peyrou, U. Pannicke, L. D. Notarangelo, F. Porta, A. R. Gennery, M. Slatter, M. J. Cowan, P. Stepensky, H. Al-Mousa, D. Al-Zahrani, SY. Pai, W. Al Herz, H. B. Gaspar, P. Veys, K. Oshima, K. Imai, H. Yabe, L. M. Noroski, N. M. Wulffraat, KW Sykora, P. Soler-Palacin, H. Muramatsu, M. Al Hilali, D. Moshous, KM. Debatin, C. Schuetz, EM Jacobsen, A. S. Schulz, K. Schwarz, A. Fischer, W. Friedrich and M. Cavazzana, on behalf of the European Society for Blood and Marrow Transplantation (EBMT) Inborn Errors Working Party

Treatment of Severe Forms of LPS-Responsive Beige-like Anchor Protein (LRBA) Deficiency by Allogeneic Hematopoietic Stem Cell Transplantation. *J Allergy Clin Immunol 2017*
M. G. Seidel, K. Böhm, F. Dogu, A. Worth, A. Thrasher, B. Florkin, A. İkincioğulları, A. Peters, S. Bakhtiar, M. Meeths, P. Stepensky, I. Meyts, S.O. Sharapova, L. Gámez-Díaz, L. Hammarström, S. Ehl, B. Grimbacher, and A. R. Gennery, on behalf of the Inborn Errors Working Party of the European Group for Blood and Marrow Transplantation

A risk factor analysis of outcomes after unrelated cord blood transplantation for children with Wiskott-Aldrich syndrome *Haematologica 2017*
Z. Shekhovtsova, C. Bonfim, A. Ruggeri, S. Nichele, K. Page, A. AlSeraihy, F. Barriga, J.Sánchez de Toledo Codina. P. Veys, JJ Boelens, K. Mellgren, H. Bittencourt, T. O'Brien, P.J. Shaw, A. Chybicka, F. Volt, F. Giannotti, E. Gluckman, J. Kurtzberg, A.R. Gennery, V. Rocha on behalf of Eurocord, Cord Blood Committee of Cellular Therapy and Immunobiology Working Party of the EBMT, Federal University of Parana, Duke University Medical Center and Inborn Errors Working Party of the EBMT

Long-term follow up of IPEX syndrome patients after different therapeutic strategies: an international multicenter retrospective study. *J Allergy Clin Immunol 2017*
F. Barzaghi, L.C. Amaya Hernandez, B. Neven, S. Ricci, Z.Y. Kucuk, J.J. Bleesing, Z. Nademi, M.A. Slatter, E.R. Ulloa, A. Shcherbina, A. Roppelt, A. Worth, J. Silva, A. Aiuti, L. Murguia-Favela, C. Speckmann, M. Carneiro-Sampaio, J. Folloni Fernandes, S. Baris, A. Ozen, E. Karakoc-Aydiner, A. Kiykim, A. Schulz, S. Steinmann, L.D. Notarangelo, E. Gambineri, P. Lionetti, W.T. Shearer, L. R. Forbes, C. Martinez, D. Moshous, S. Blanche, A. Fisher, F.M. Ruemmele, C. Tissandier, M. Ouachee-Chardin, F. Rieux-Laucat, M. Cavazzana, W. Qasim, B. Lucarelli, M.H. Albert, I. Kobayashi, L. Alonso, C. Diaz De Heredia, H. Kanegane, A. Lawitschka, JJ. Seo, M. Gonzalez-Vicent, M.A. Diaz, R.K. Goyal, M.G. Sauer, A. Yesilipek, M. Kim, Y. Yilmaz-Demirdag, M. Bhatia, J. Khlevner, E.J. Richmond Padilla, S. Martino, D. Montin, O. Neth, A. Molinos-Quintana, J. Valverde-Fernandez, A. Broides, V. Pinsk, A. Ballauf, F. Haerynck, V. Bordon, C. Dhooge, M.L. Garcia-Lloret, R. G. Bredius, K. Kałwak, E.Haddad, M.G. Seidel, G. Duckers, SY Pai, C.C. Dvorak, S. Ehl, F. Locatelli, F. Goldman, A.R. Gennery, M. J. Cowan, MG Roncarolo, and R. Bacchetta on behalf of the Primary Immune Deficiency Treatment Consortium (PIDTC) and the Inborn Errors Working Party (IEWP) of the European Society for Blood and Marrow Transplantation (EBMT)

Significant Collaborations

ESID · SCETIDE · eurocord · CIBMTR (Center for International Blood & Marrow Transplant Research) · PIDTC · cell pid

EBMT Registry

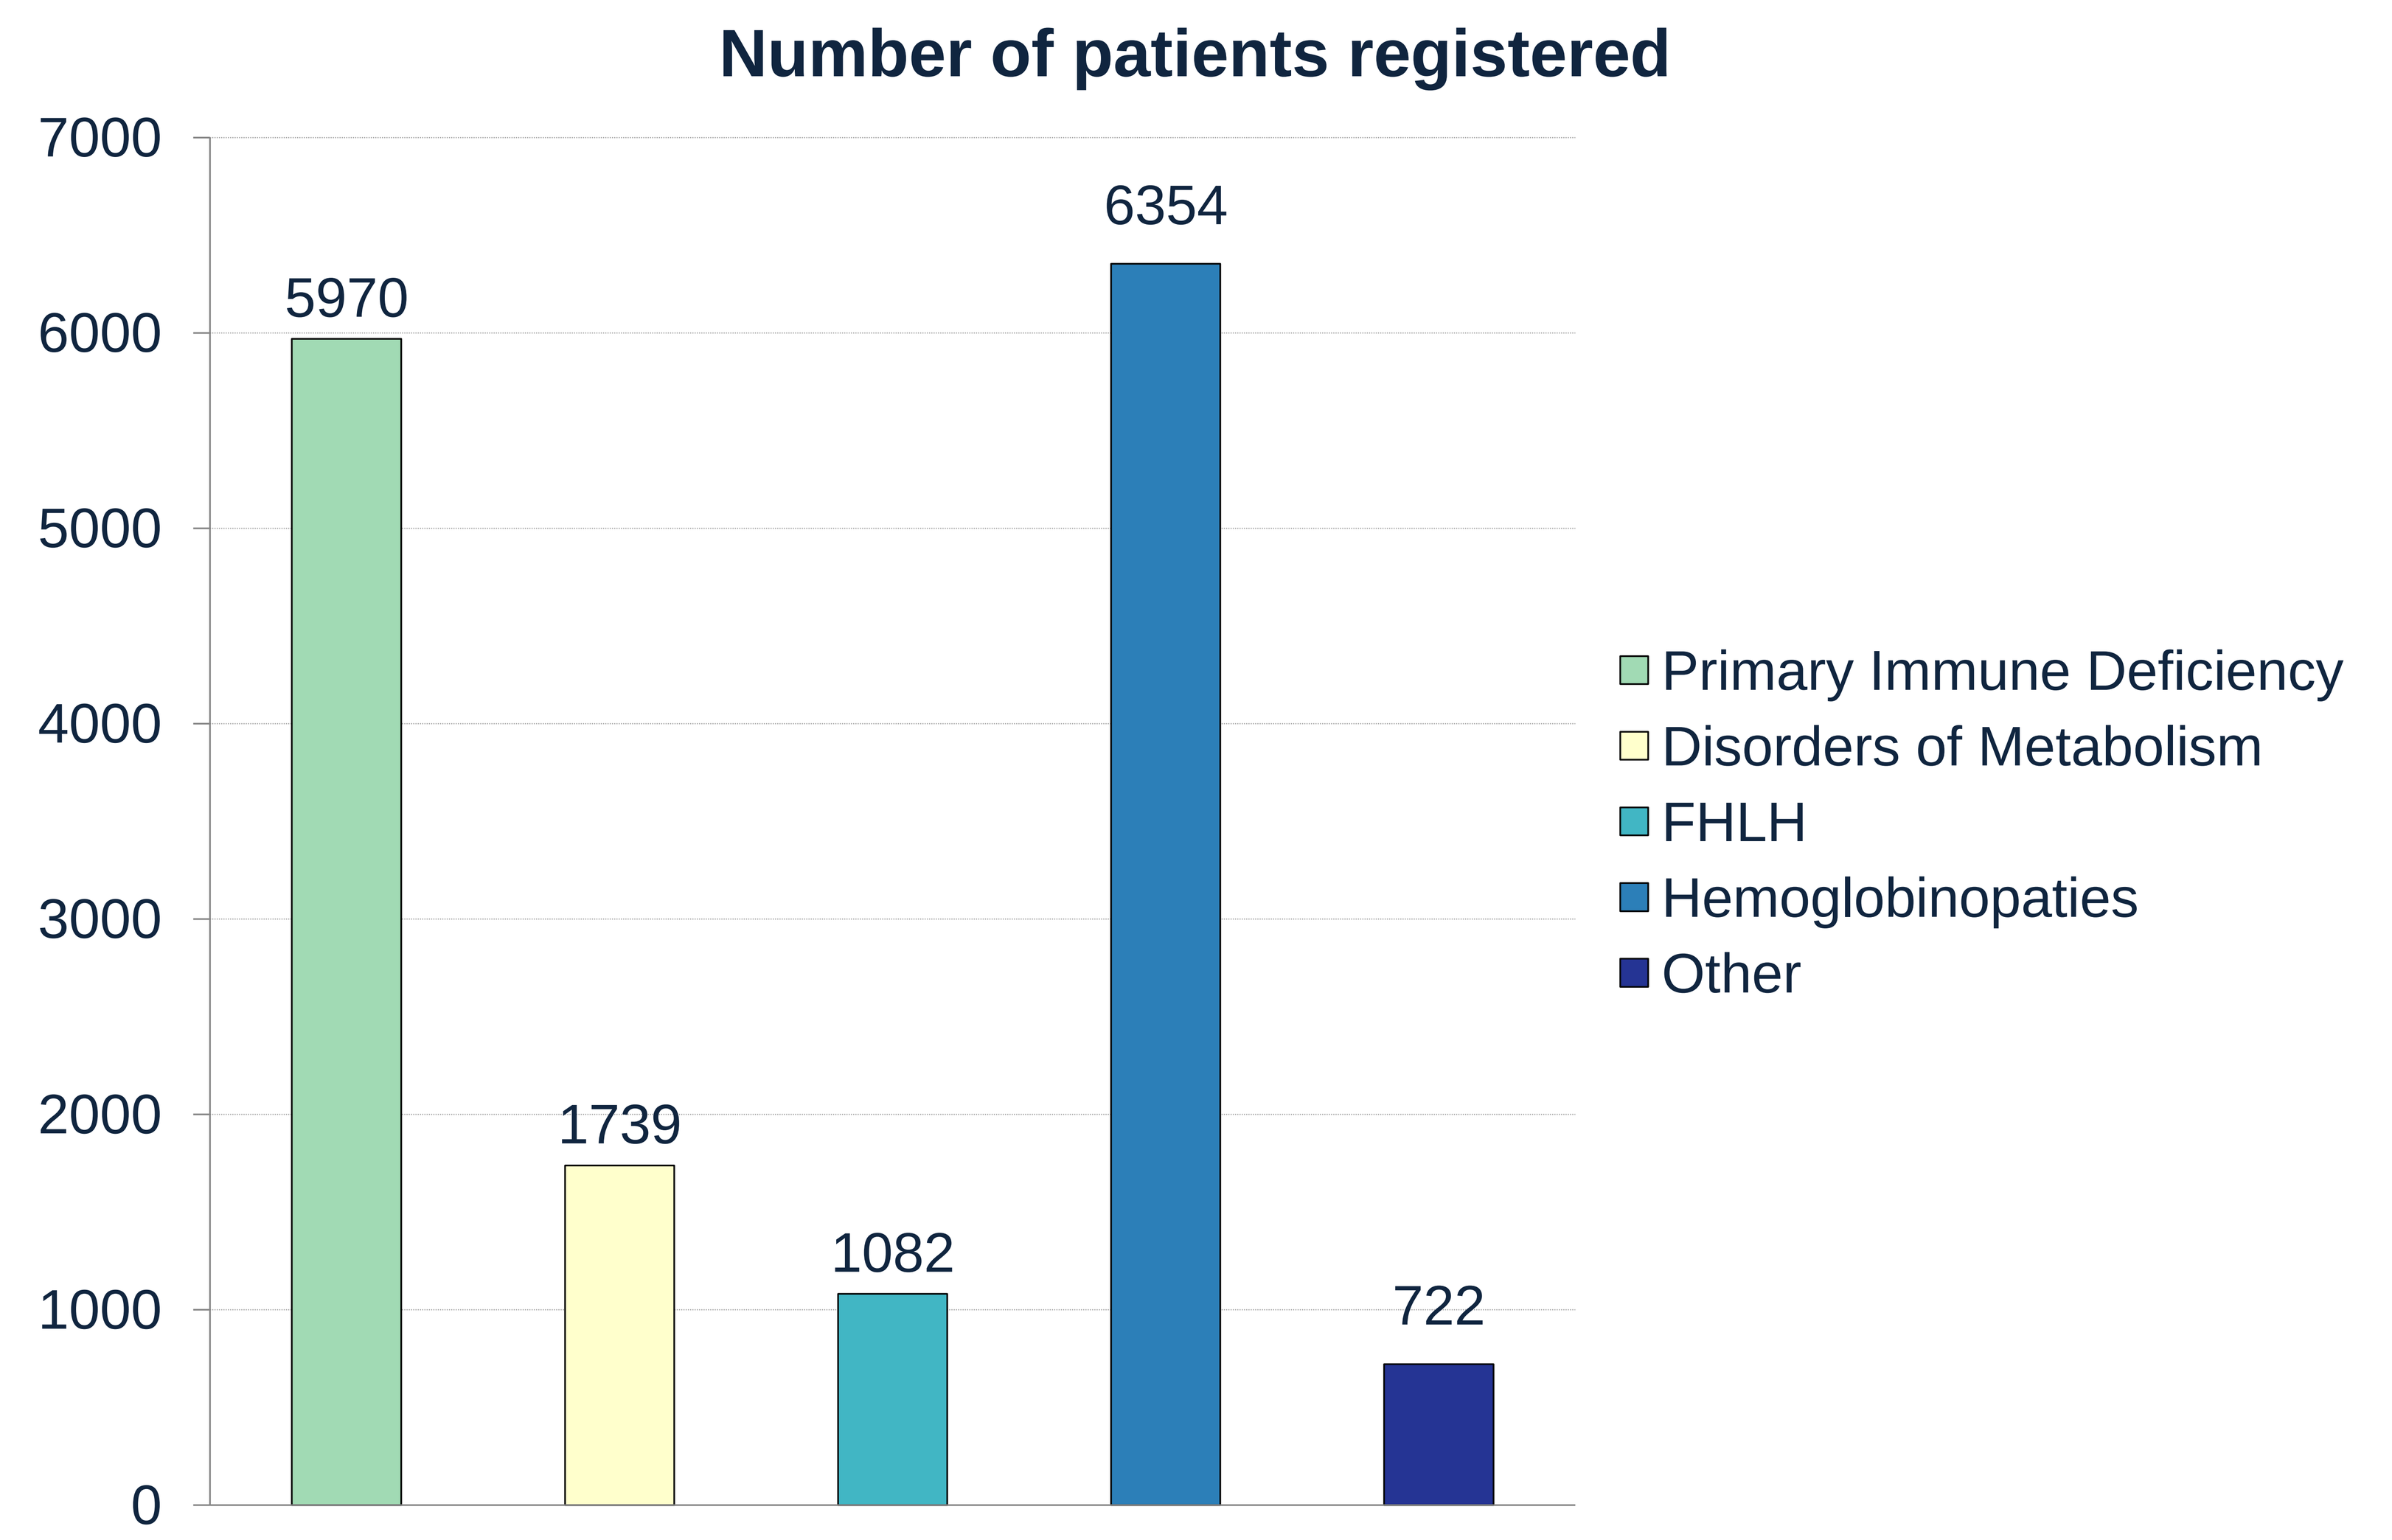
Number of patients registered
7000
6000
5000
4000
3000
2000
1000
0
5970
1739
1082
6354
722
Primary Immune Deficiency
Disorders of Metabolism
FHLH
Hemoglobinopaties
Other

Lisbon IEWP-events

Monday March 19th
IEWP Business meeting
7.30 – 9.00
Room 5 A

Tuesday March 20th
IEWP session
11.00 – 12.30
Auditorium V

Program IEWP session:

- 11:00-11:18 Gene Therapy vs SCT for ADA-SCID
 Bobby Gaspar (UK)
- 11:18-11:36 SCT in leukocyte adhesion deficiency (IEWP/PDWP study)
 Shahrzad Bakhtiar (Germany)
- 11:36-11:54 HaploSCT in Hemoglobinopathies
 Josu de la Fuente (UK)
- 11:54-12:12 Gene therapy in Hemoglobinopathies
 Alessandro Aiuti (Italy)
- 12:12-12:30 Outcomes after CBT in IMD (IEWP/EUROCORD study)
 Su Han Lum (UK)

Future Events 2018

PDWP, IDWP, IEWP & Nurses group meeting
Verona June 7-9, 2018

ESID meeting
Lisbon 24-27 October, 2018

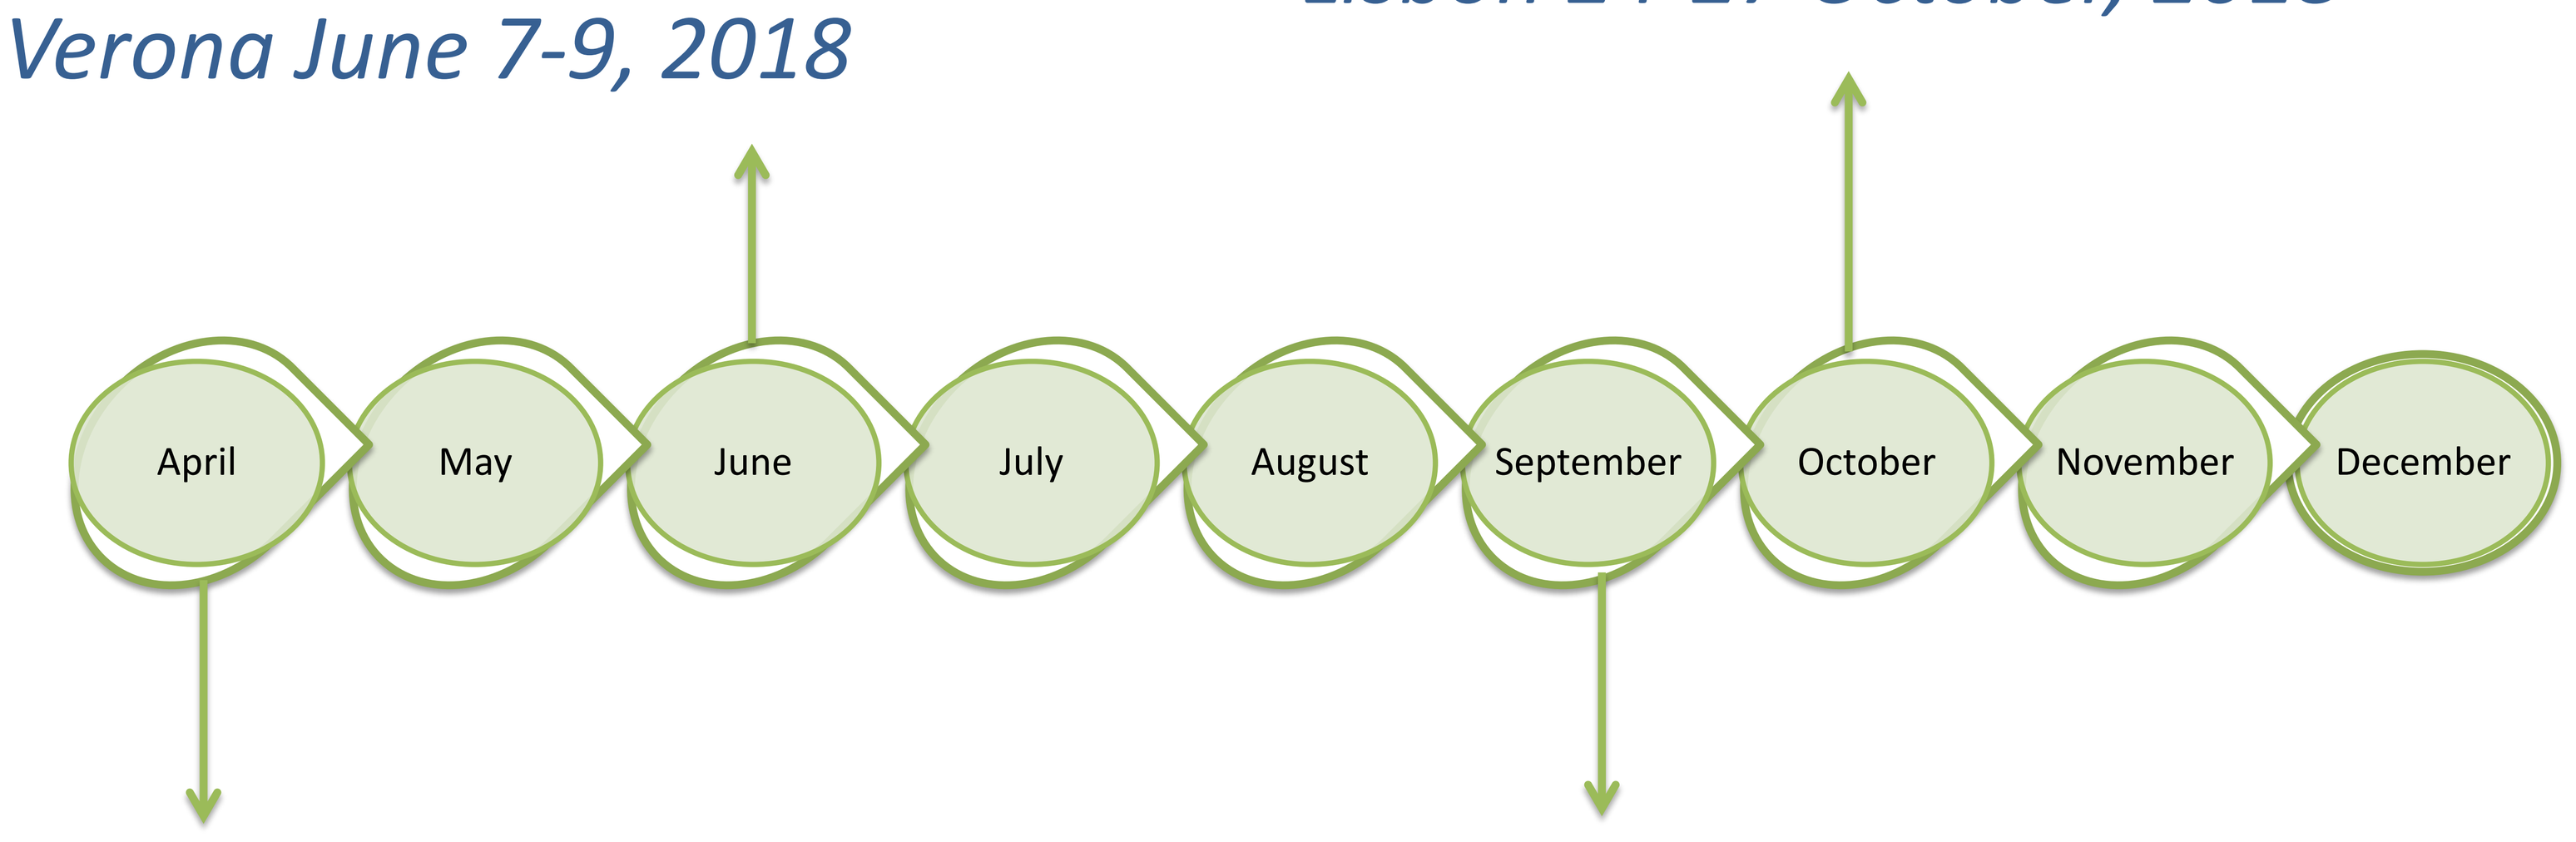
April
May
June
July
August
September
October
November
December

Workshop on HSCT for PID in adults and young adolescents (AYA)
Munich April 13th, 2018

IEWP autumn meeting
Leiden 28-30 September, 2018

Contact IEWP

Arjan Lankester	Chair	A.Lankester@lumc.nl
Yvonne Wijkhuisen	Secretary A. Lankester	Y.W.Wijkhuisen-Helvenstein@lumc.nl
Michael Albert	Secretary/Co-Chair	Michael.Albert@med.uni-muenchen.de
Henric-Jan Blok	EBMT Data manager	H.J.P.Blok@lumc.nl
Sheree Hazelaar	EBMT jr. Data manager	S.Hazelaar@lumc.nl

#EBMT18
12
www.ebmt.org